Sex disparity in response of murine sclerosing cholangitis to short-term treatment with FXR agonists

Tiffany Shi[1], Ana Catalina Arce-Clachar[1], Mary Mullen[1], Alvaro Ortiz[3], Kyoung-Jin Lee[3], Kumar Shanmukhappa[2], Wujuan Zhang[2], Brandee Wagner[3], Alexander Miethke[1]
[1]Division of Gastroenterology, Hepatology and Nutrition, Cincinnati Children's Hospital Medical Center, Cincinnati, OH, USA.
[2]Department of Pathology and Laboratory Medicine, Cincinnati Children's Hospital Medical Center, Cincinnati, OH, USA.
[3]Metacrine, Inc. San Diego, CA, USA.

Cincinnati Children's
changing the outcome together

Introduction

Primary sclerosing cholangitis (PSC) is a progressive fibrosing cholangiopathy where patients develop severe cholestasis and require liver transplantation for prolonged survival. PSC is characterized by chronic inflammation, increased bile duct proliferation and fibrosis leading to portal hypertension and liver failure. There is also a gender disparity in which PSC is more prevalent in males than in females. Currently, PSC lacks medical therapies.

Bile acids (BAs) are drivers of progressive liver injury in fibrosing cholangiopathies like PSC. Response to anti-cholestatic therapy also shows a sex predilection in PSC. Predictors for response to anti-cholestatic therapy in these conditions are largely undefined.

The MDR2-/- mouse is an established model for the biliary fibrosis accompanying PSC.

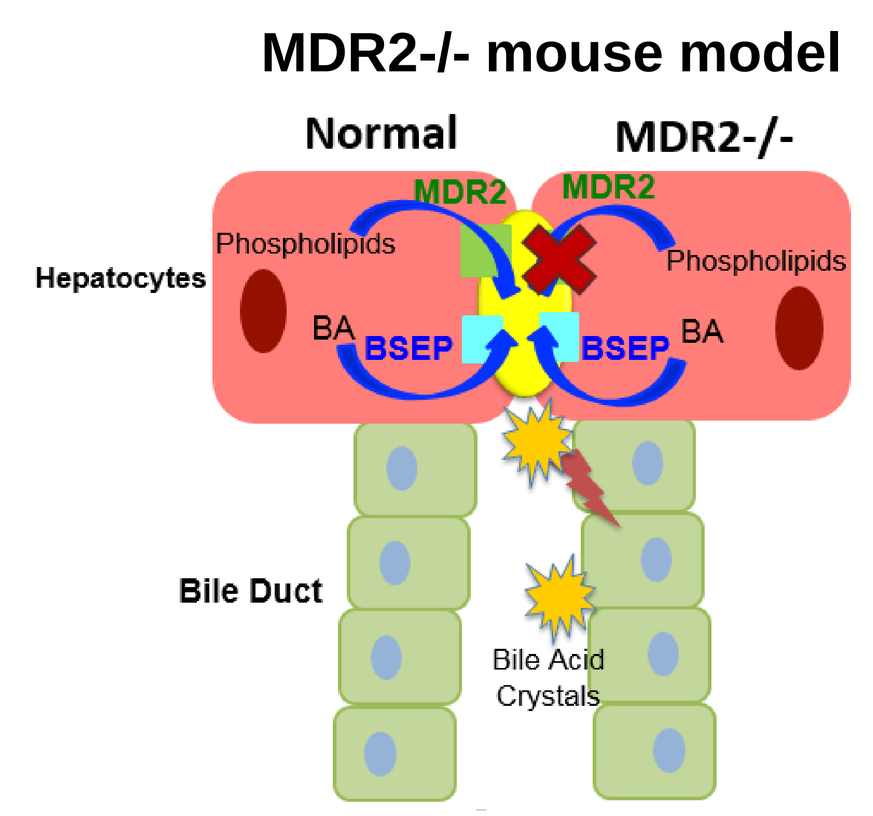
MDR2-/- mouse model
Normal
MDR2-/-
Hepatocytes
MDR2
Phospholipids
BA
BSEP
Bile Duct
Bile Acid Crystals

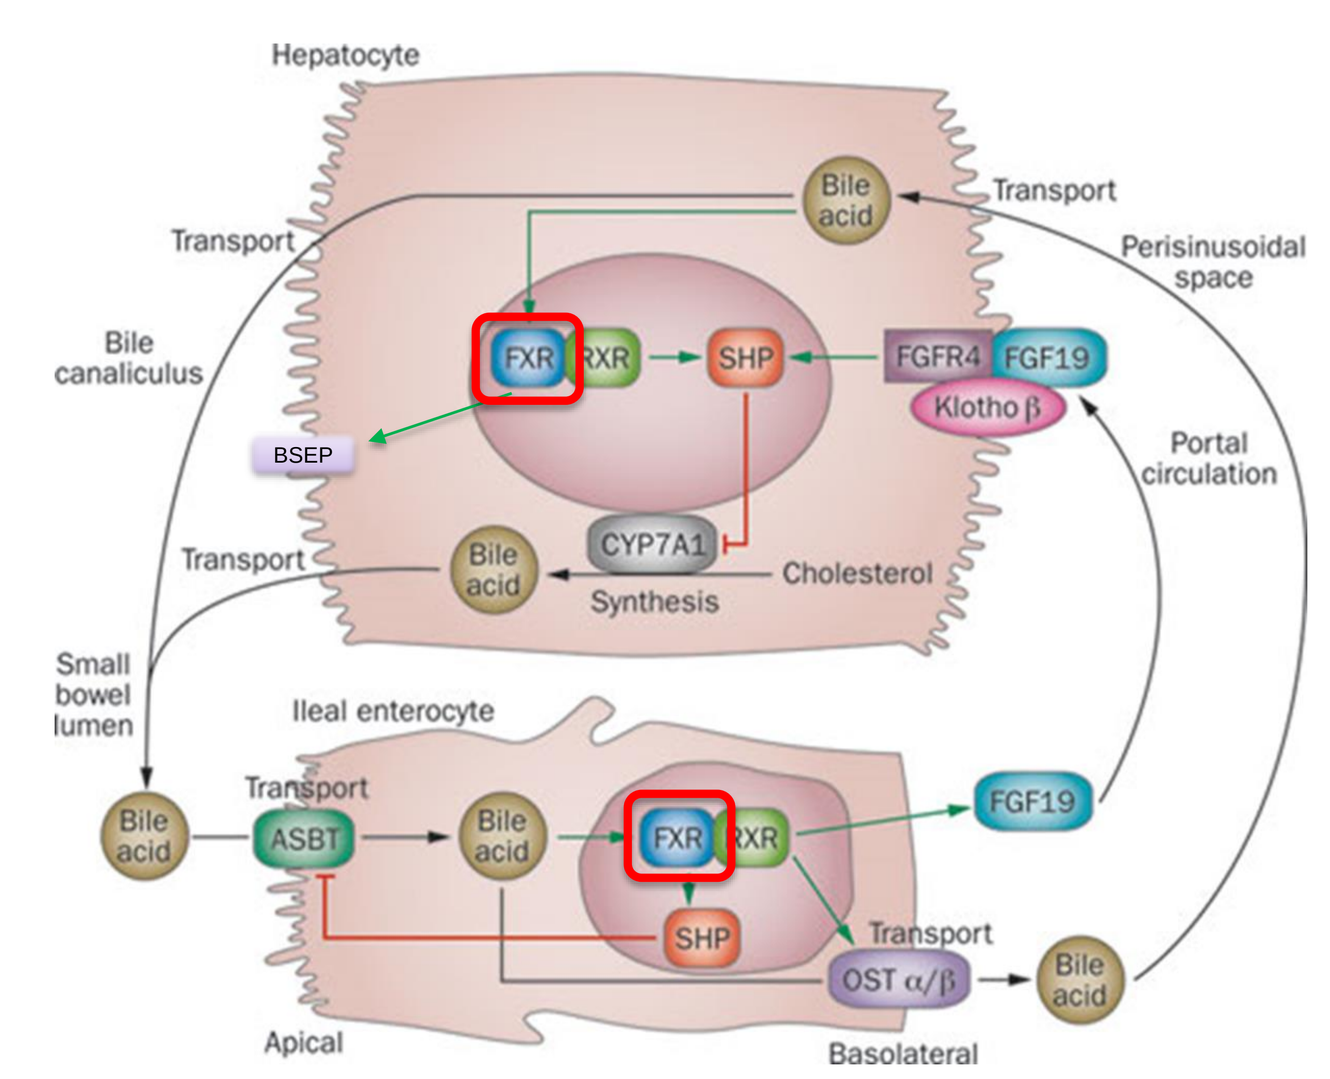
Hepatocyte
Transport
Bile acid
Perisinusoidal space
Bile canaliculus
FXR
RXR
SHP
FGFR4
FGF19
Klotho β
BSEP
Portal circulation
CYP7A1
Cholesterol
Synthesis
Small bowel lumen
Ileal enterocyte
ASBT
OST α/β
Apical
Basolateral

Adapted from Rao, et al., *Gastroenterology*, 2010

BAs circulate through the body via the enterohepatic circulation. After synthesis in the liver, BAs are exported via BSEP into the small intestine, and are reabsorbed in the terminal small intestine via ASBT. BAs activate FXR, which signals through SHP to upregulate FGF15, which travels through the portal circulation and activates SHP in the liver via FGFR4. Liver SHP suppresses BA synthesis. BAs are also circulated back to the liver where it can act upon liver FXR.

Hypothesis

Short-term treatment with FXR agonist M345 will attenuate MDR2-/- sclerosing cholangitis phenotype in gender specific manner.
Aim 1: Determine sex specific change in SC phenotype following treatment with FXR agonist
Aim 2: Explore sex differences in FXR-induced hepatic and intestinal gene expression

Experimental Design

MDR2 -/- and WT Balb/c
(gift from Prof. Lammert, Homburg)

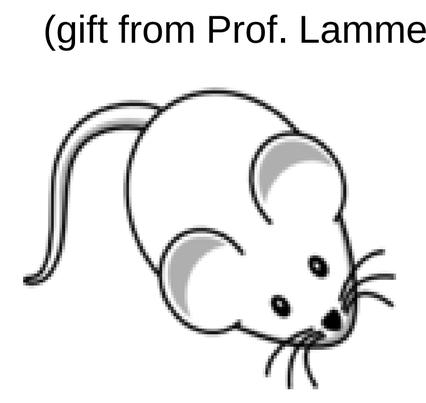

Start on breeder chow 5058

Day of Life

30 **45**

Harvest
- Blood (plasma, serum)
- Liver → Histology, BA

MDR2 +/- & -/- Balb/c
Starting DOL45

1. M345 (30mg/kg/day)- a fexaramine derivative FXR agonist provided by Metacrine
2. Control Corn Oil

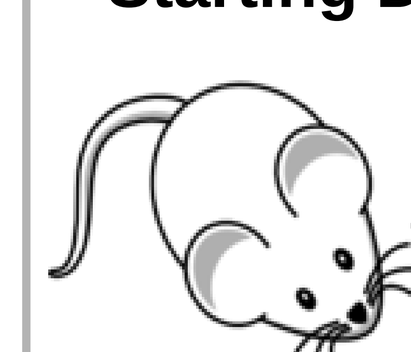

Day of Treatment

- 16 hrs 1 2 3 4 5 6 7 +8 hrs

- Start on breeder chow 5058

Daily oral gavage
- Total 8 doses, 7 days

Harvest
- Blood (plasma, serum, PV)
- Bile
- Liver → RNA, Histology, BA
- Intestine → RNA

Results

Figure 1: Serum liver biochemistries at baseline

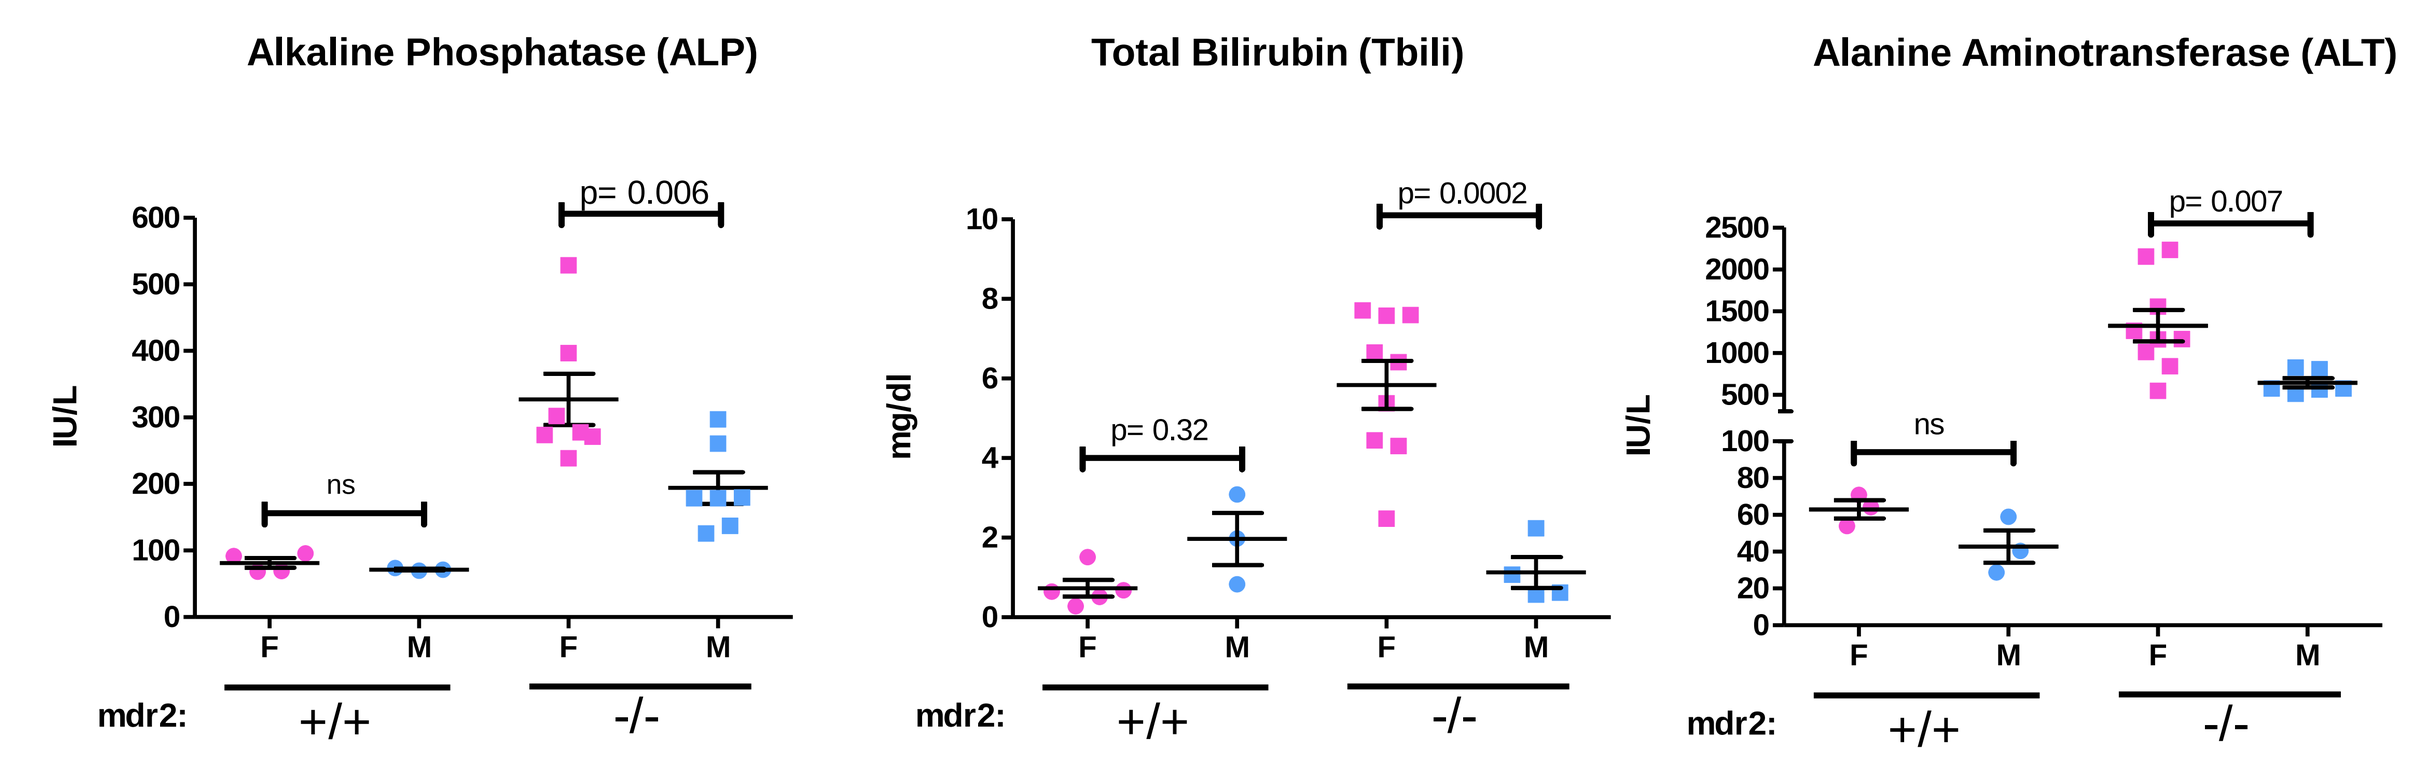
Alkaline Phosphatase (ALP)
Total Bilirubin (Tbili)
Alanine Aminotransferase (ALT)
IU/L
mg/dl
ns
p= 0.006
p= 0.32
p= 0.0002
p= 0.007
F M F M
mdr2: +/+ -/-

Figure 2a: Liver histopathology at baseline

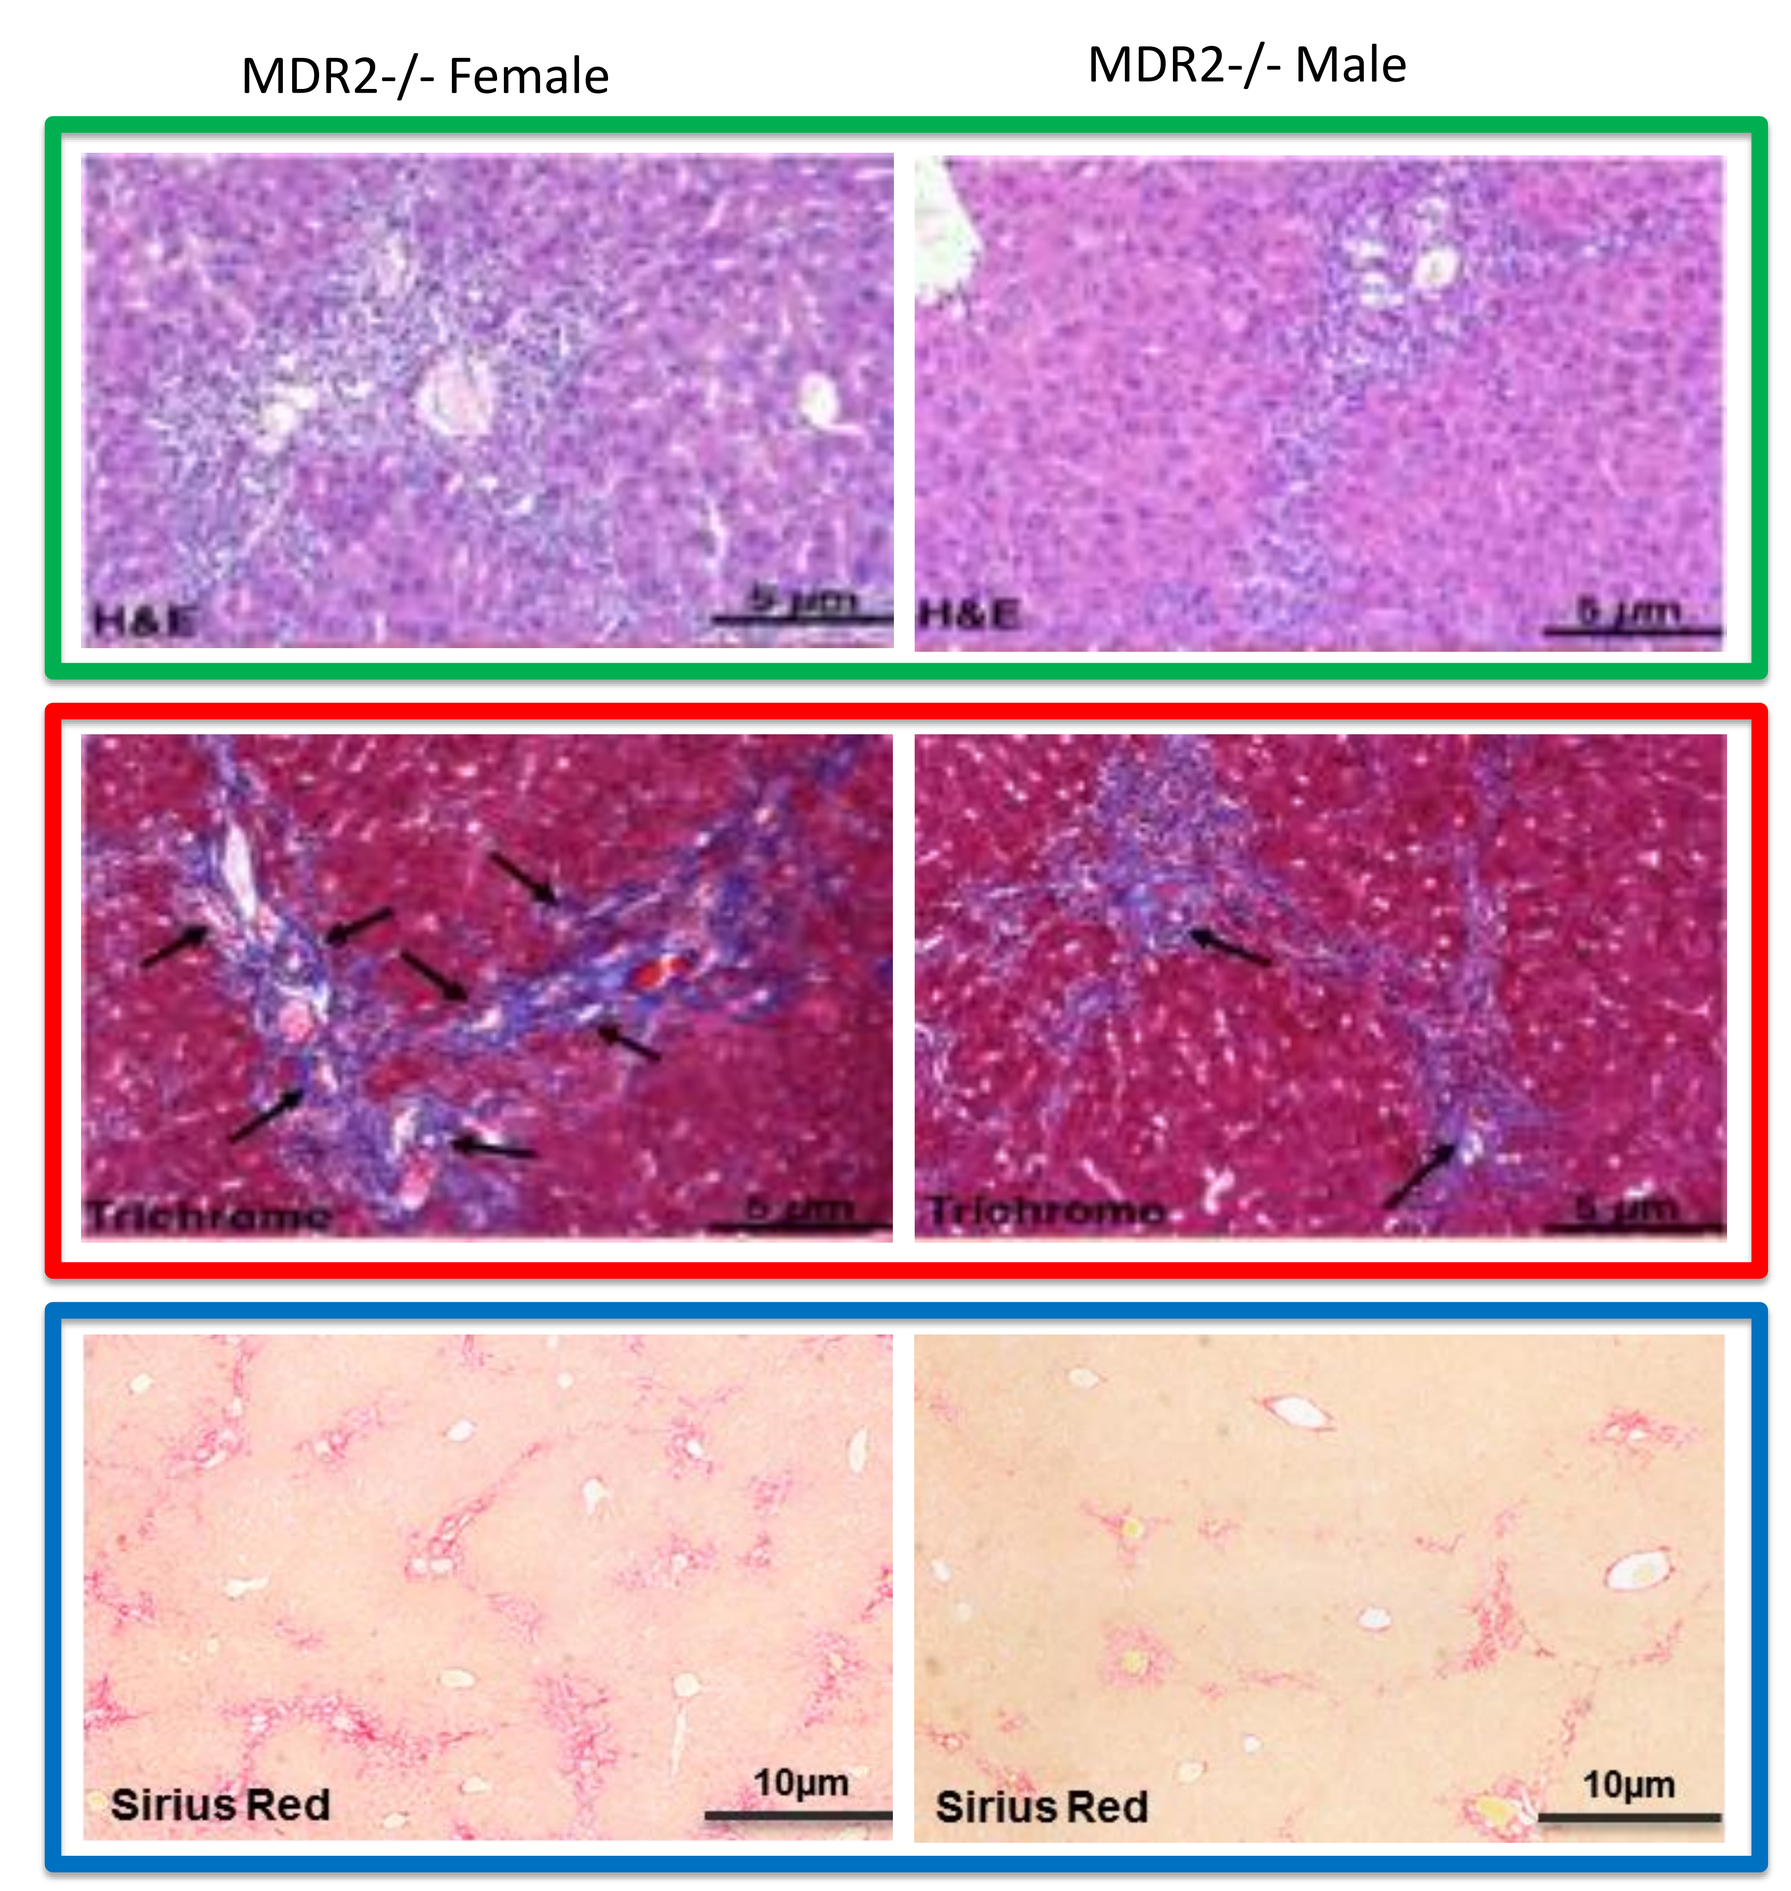
MDR2-/- Female
MDR2-/- Male
H&E
Trichrome
Sirius Red
10µm

Figure 2b: Blinded review of histopathology and Sirius red image analysis

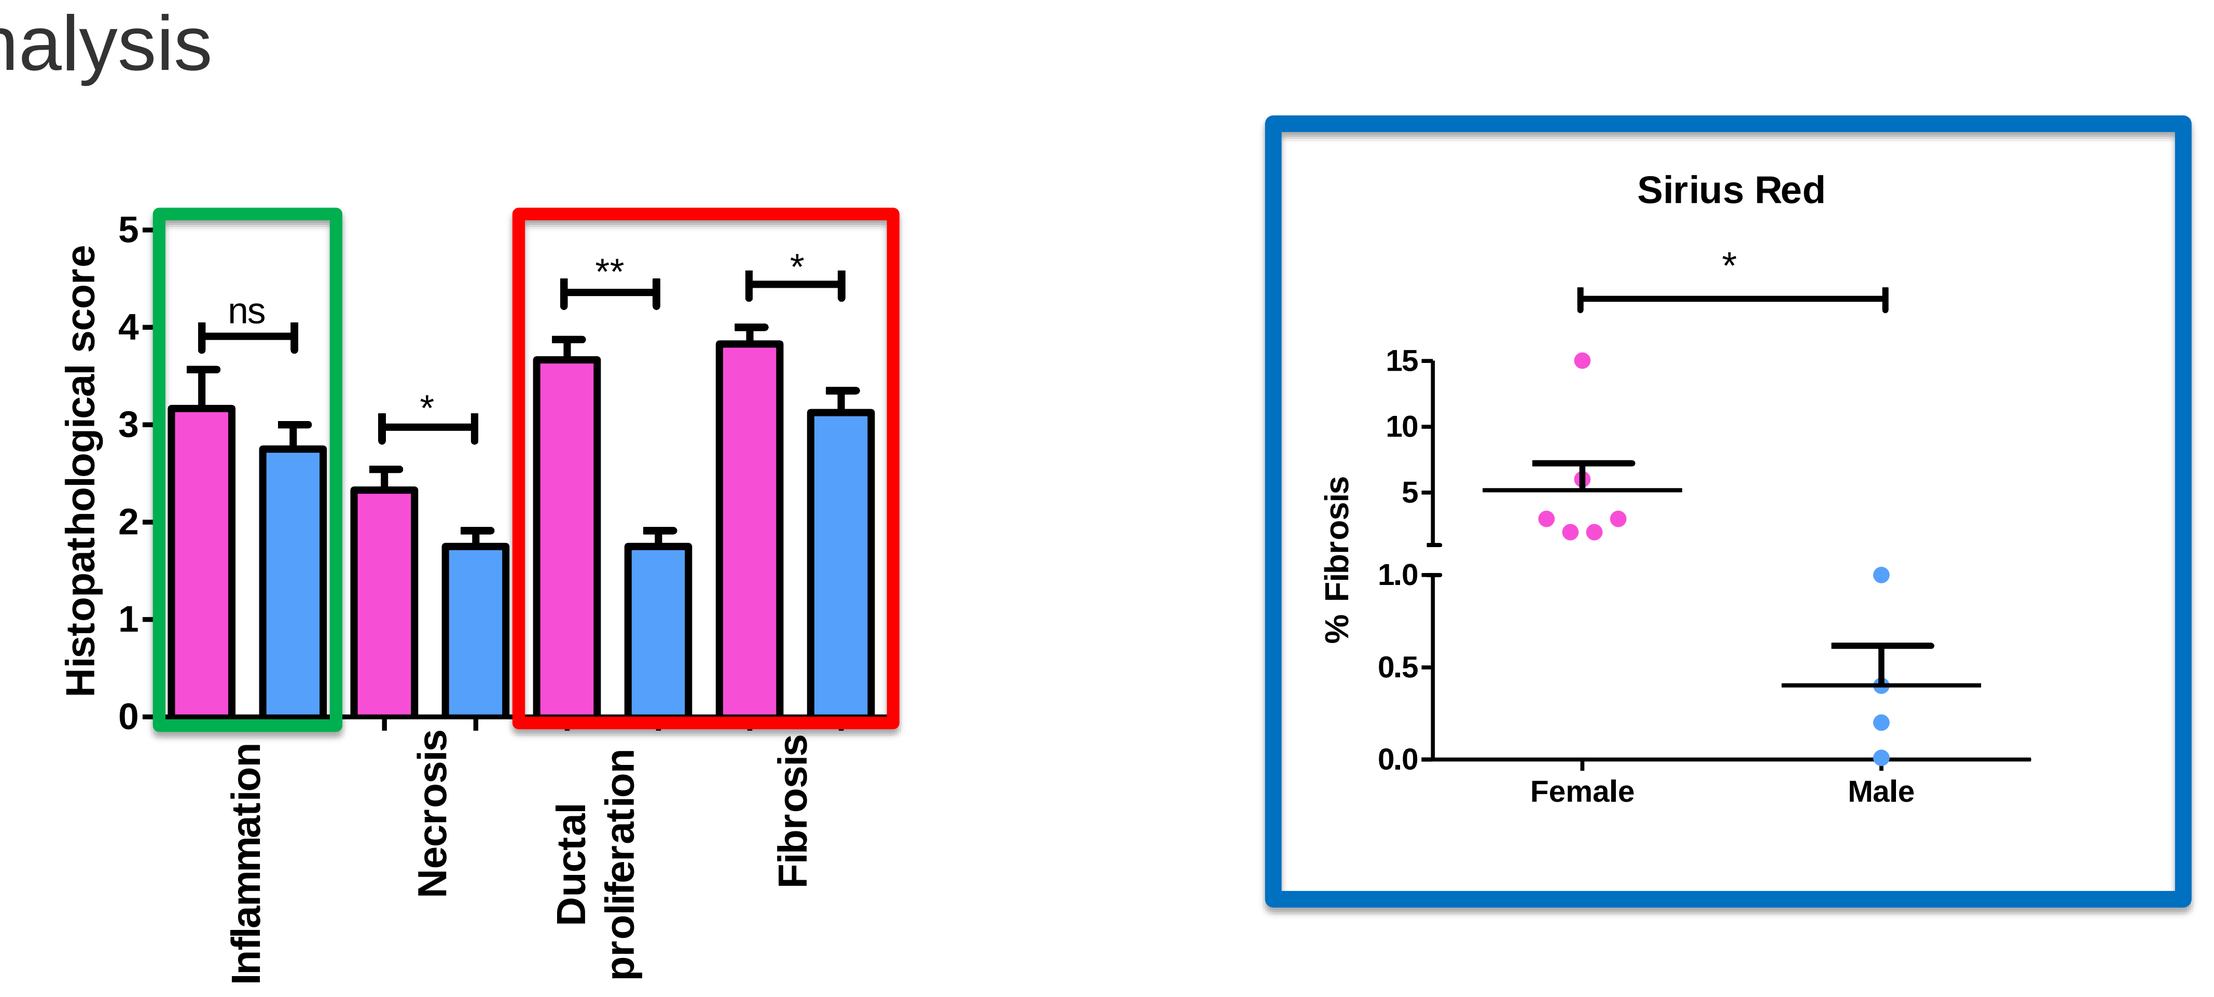
Histopathological score
ns
Inflammation
Necrosis
Ductal proliferation
Fibrosis
Sirius Red
% Fibrosis
Female
Male

Results

Figure 3: Weight changes from baseline upon 8-day treatment with M345

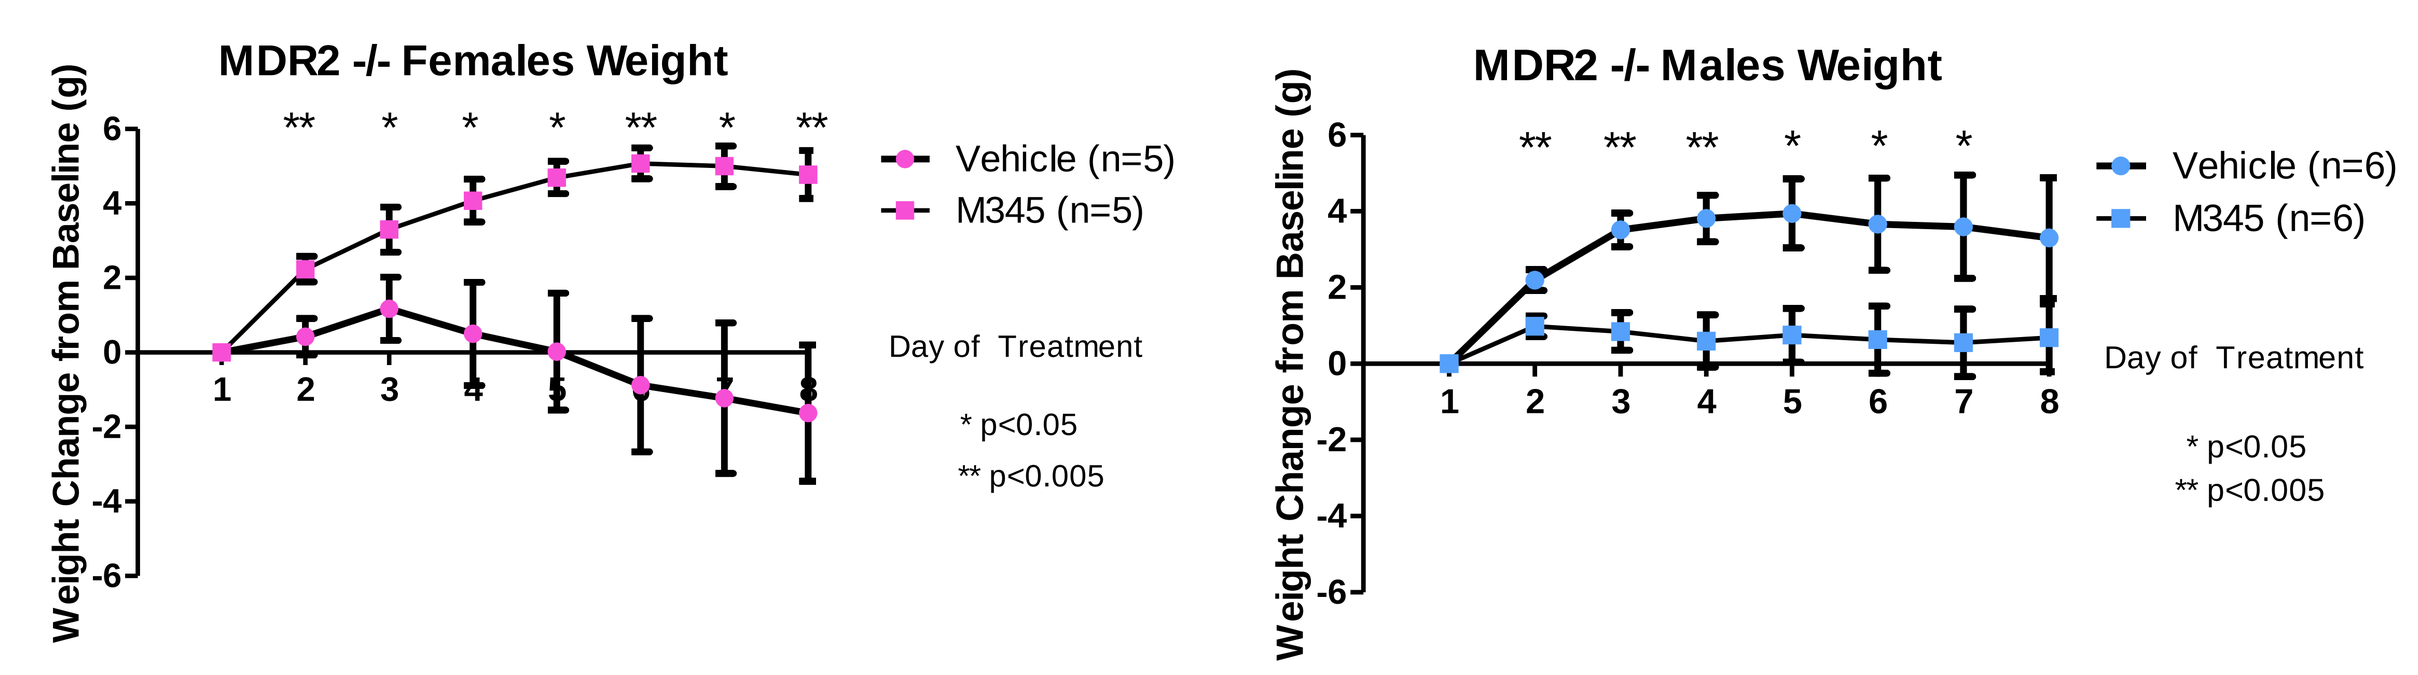
MDR2 -/- Females Weight
MDR2 -/- Males Weight
Weight Change from Baseline (g)
Vehicle (n=5)
M345 (n=5)
Vehicle (n=6)
M345 (n=6)
Day of Treatment
* p<0.05
** p<0.005

Figure 4: Effects of short-term M345 treatment on serum liver biochemistries

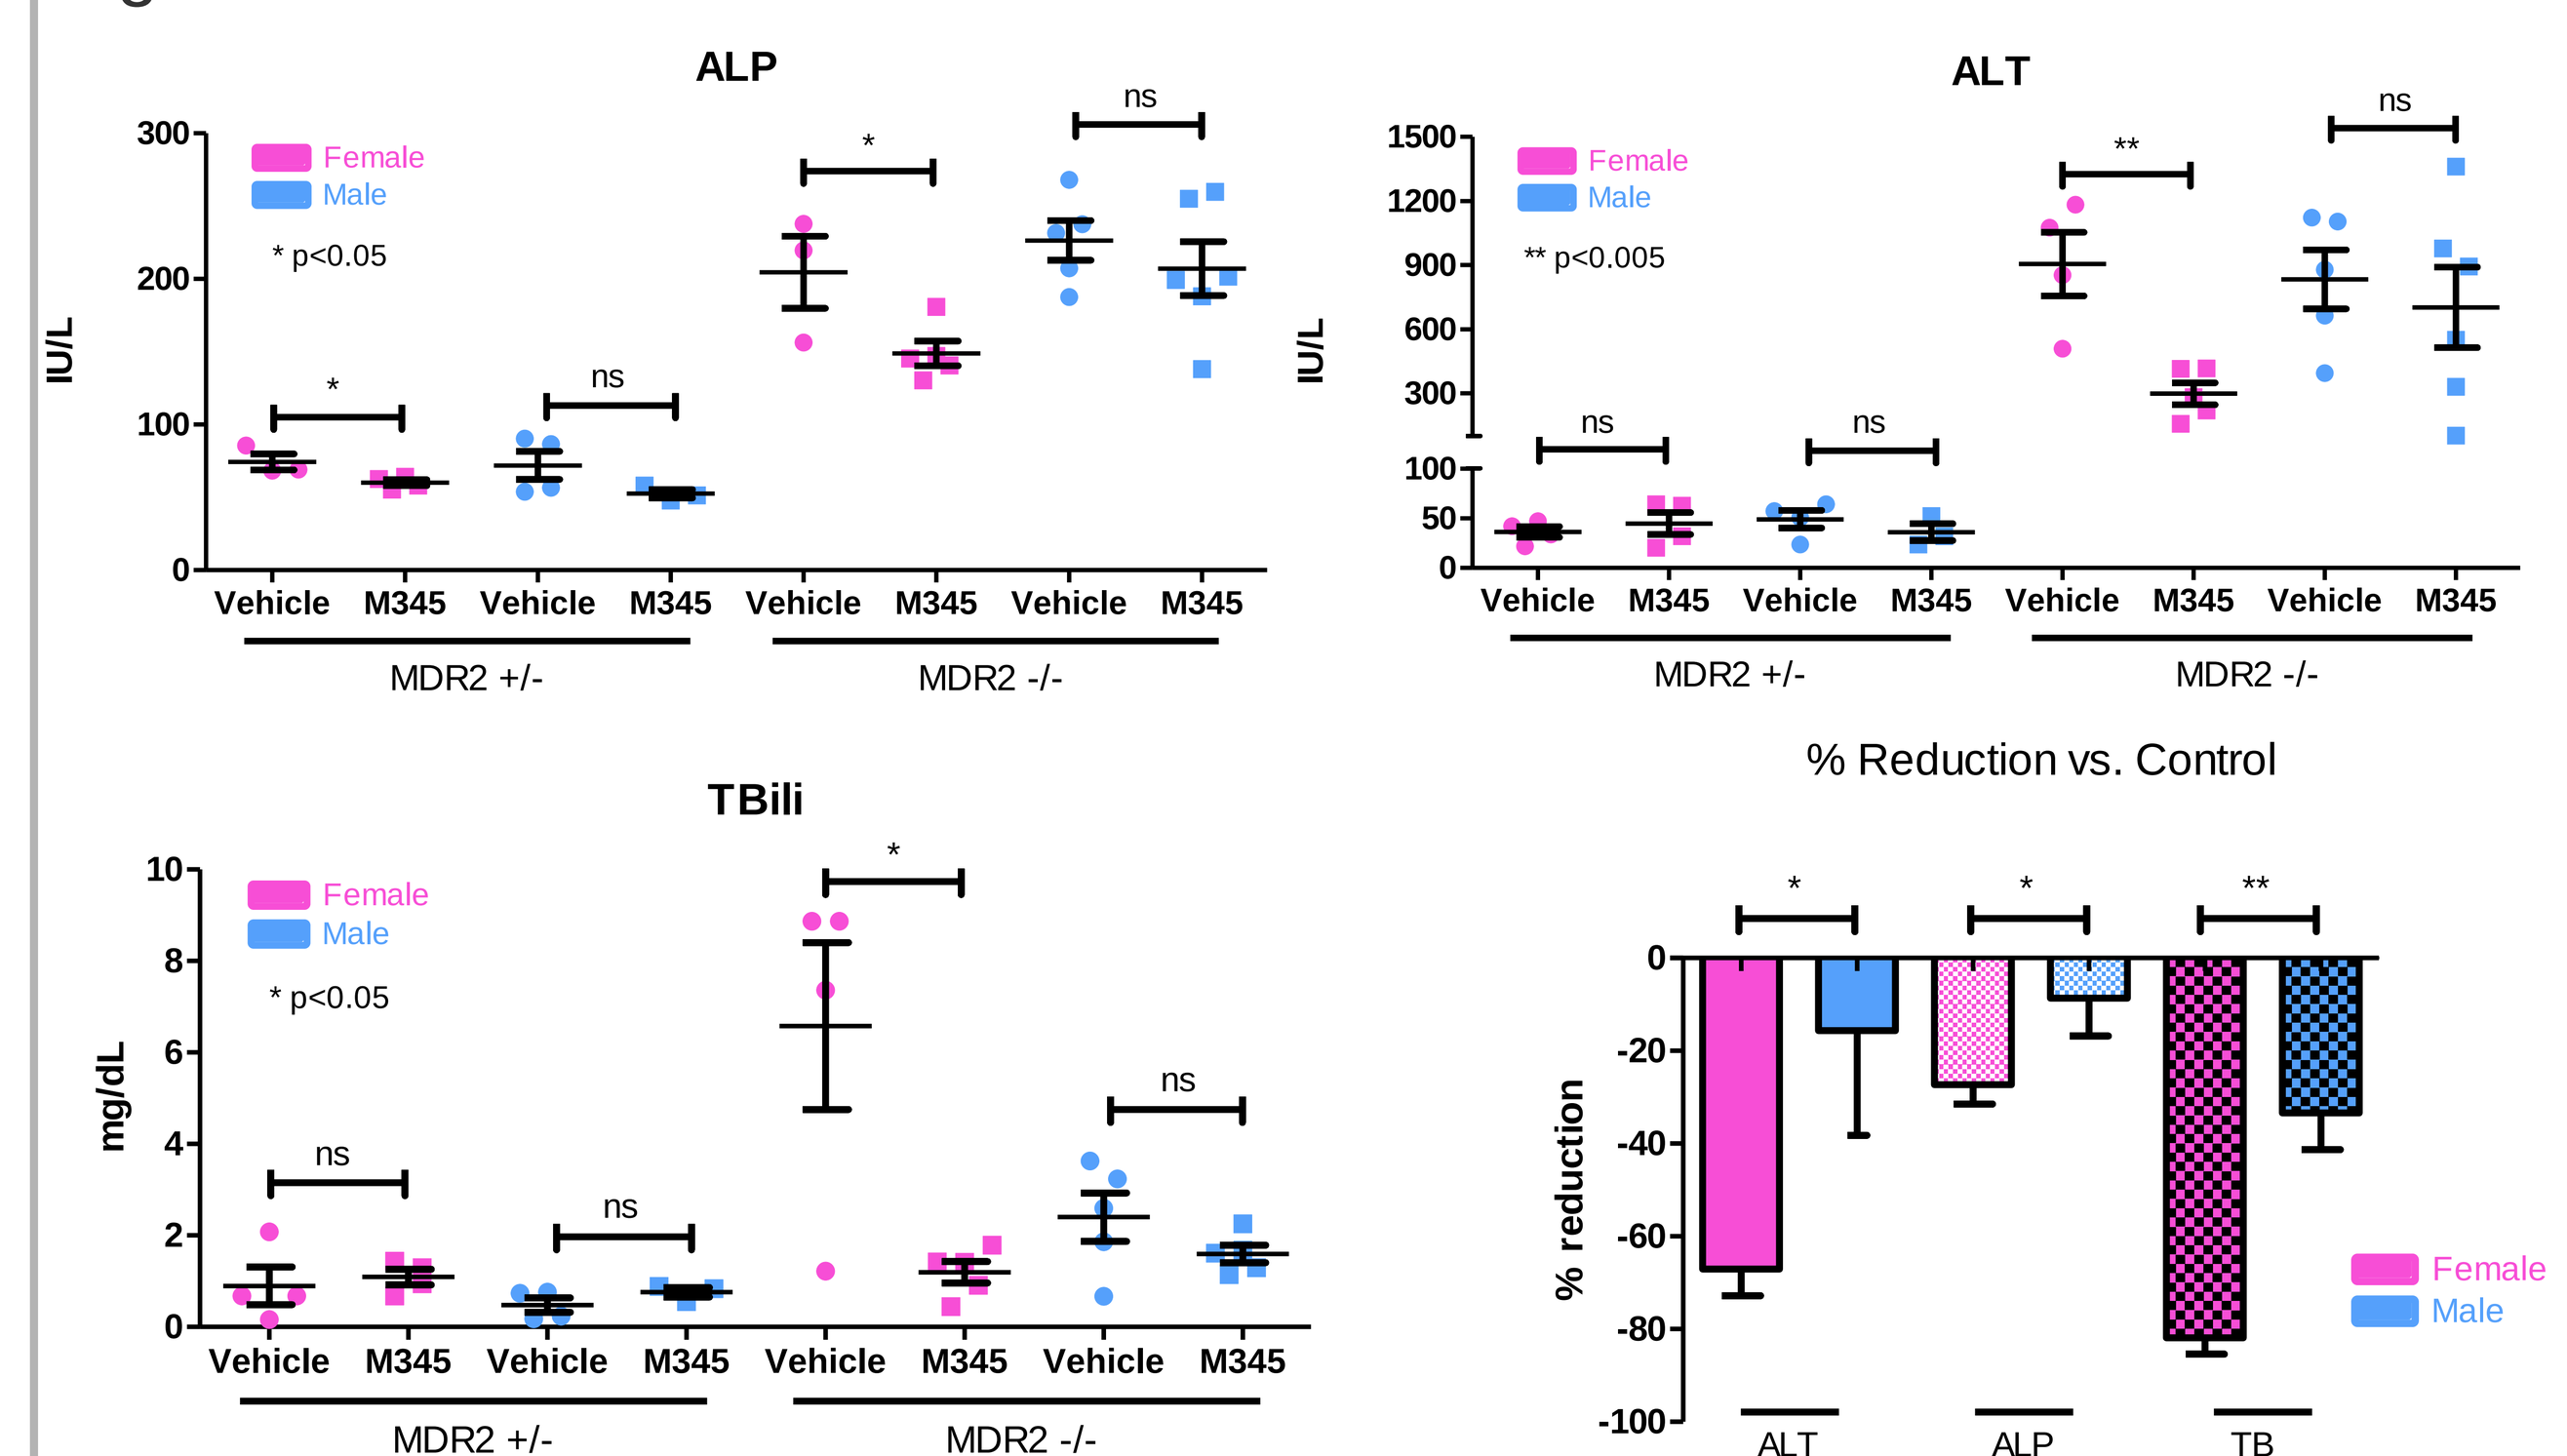
ALP
ALT
TBili
Female
Male
* p<0.05
** p<0.005
IU/L
mg/dL
Vehicle M345 Vehicle M345 Vehicle M345 Vehicle M345
MDR2 +/-
MDR2 -/-
% Reduction vs. Control
% reduction
ALT
ALP
TB

Figure 5: Effect of short-term M345 treatment on liver TNF-α gene expression

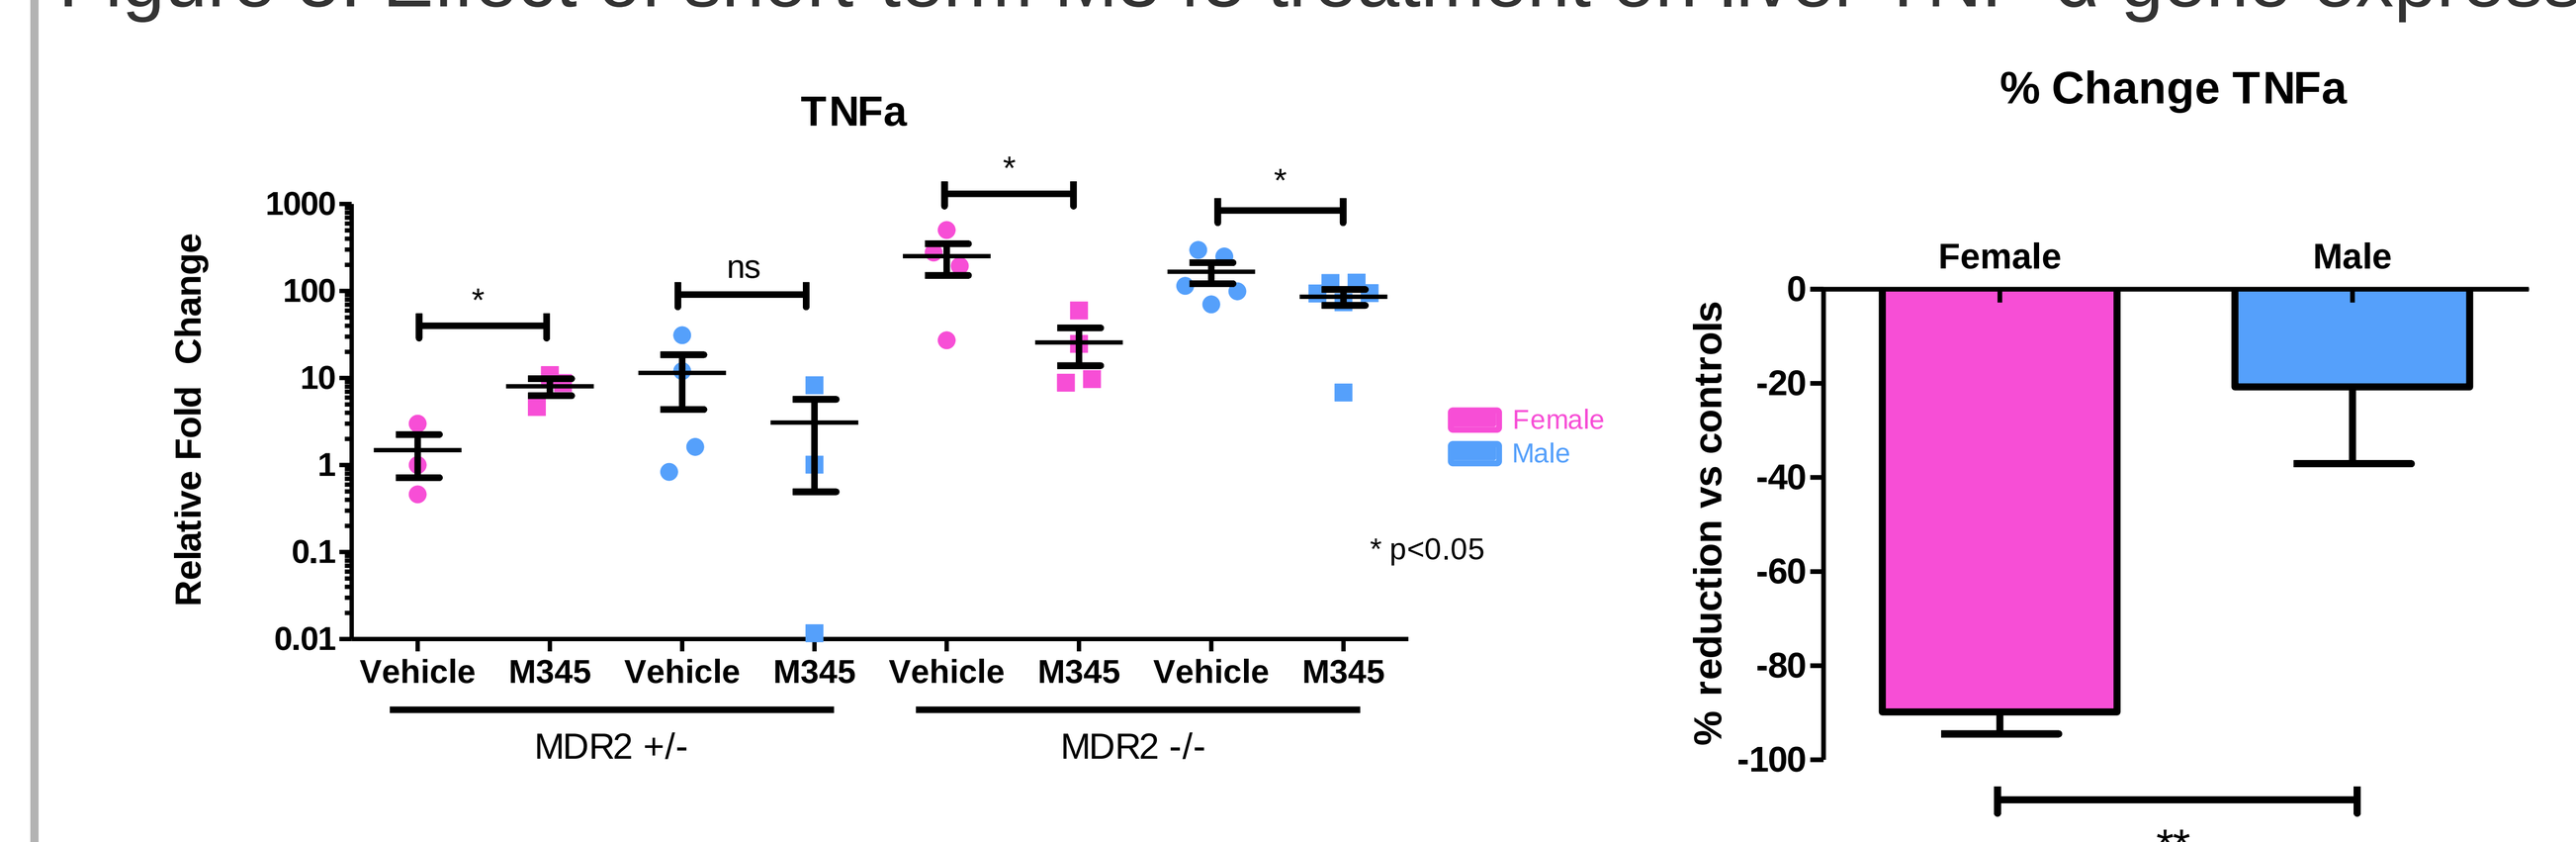
TNFa
Relative Fold Change
% Change TNFa
% reduction vs controls
Female
Male
* p<0.05
MDR2 +/-
MDR2 -/-

Figure 6: Hepatic concentration of M345

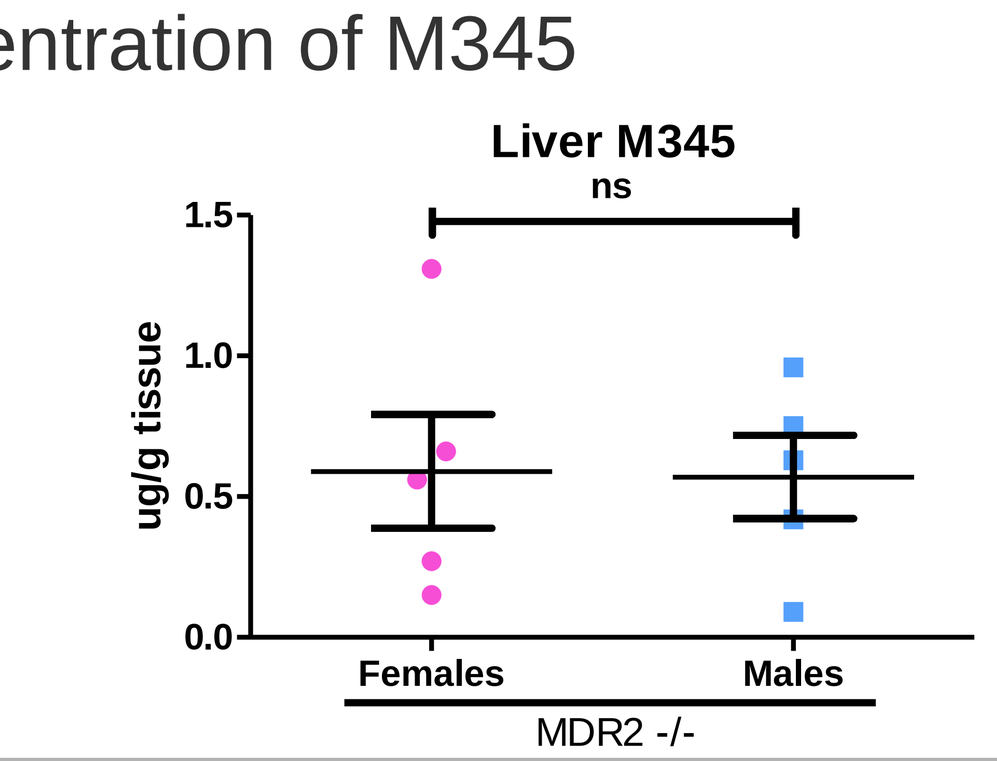
Liver M345
ns
ug/g tissue
Females
Males
MDR2 -/-

Figure 7: Liver response to 8-day M345 treatment

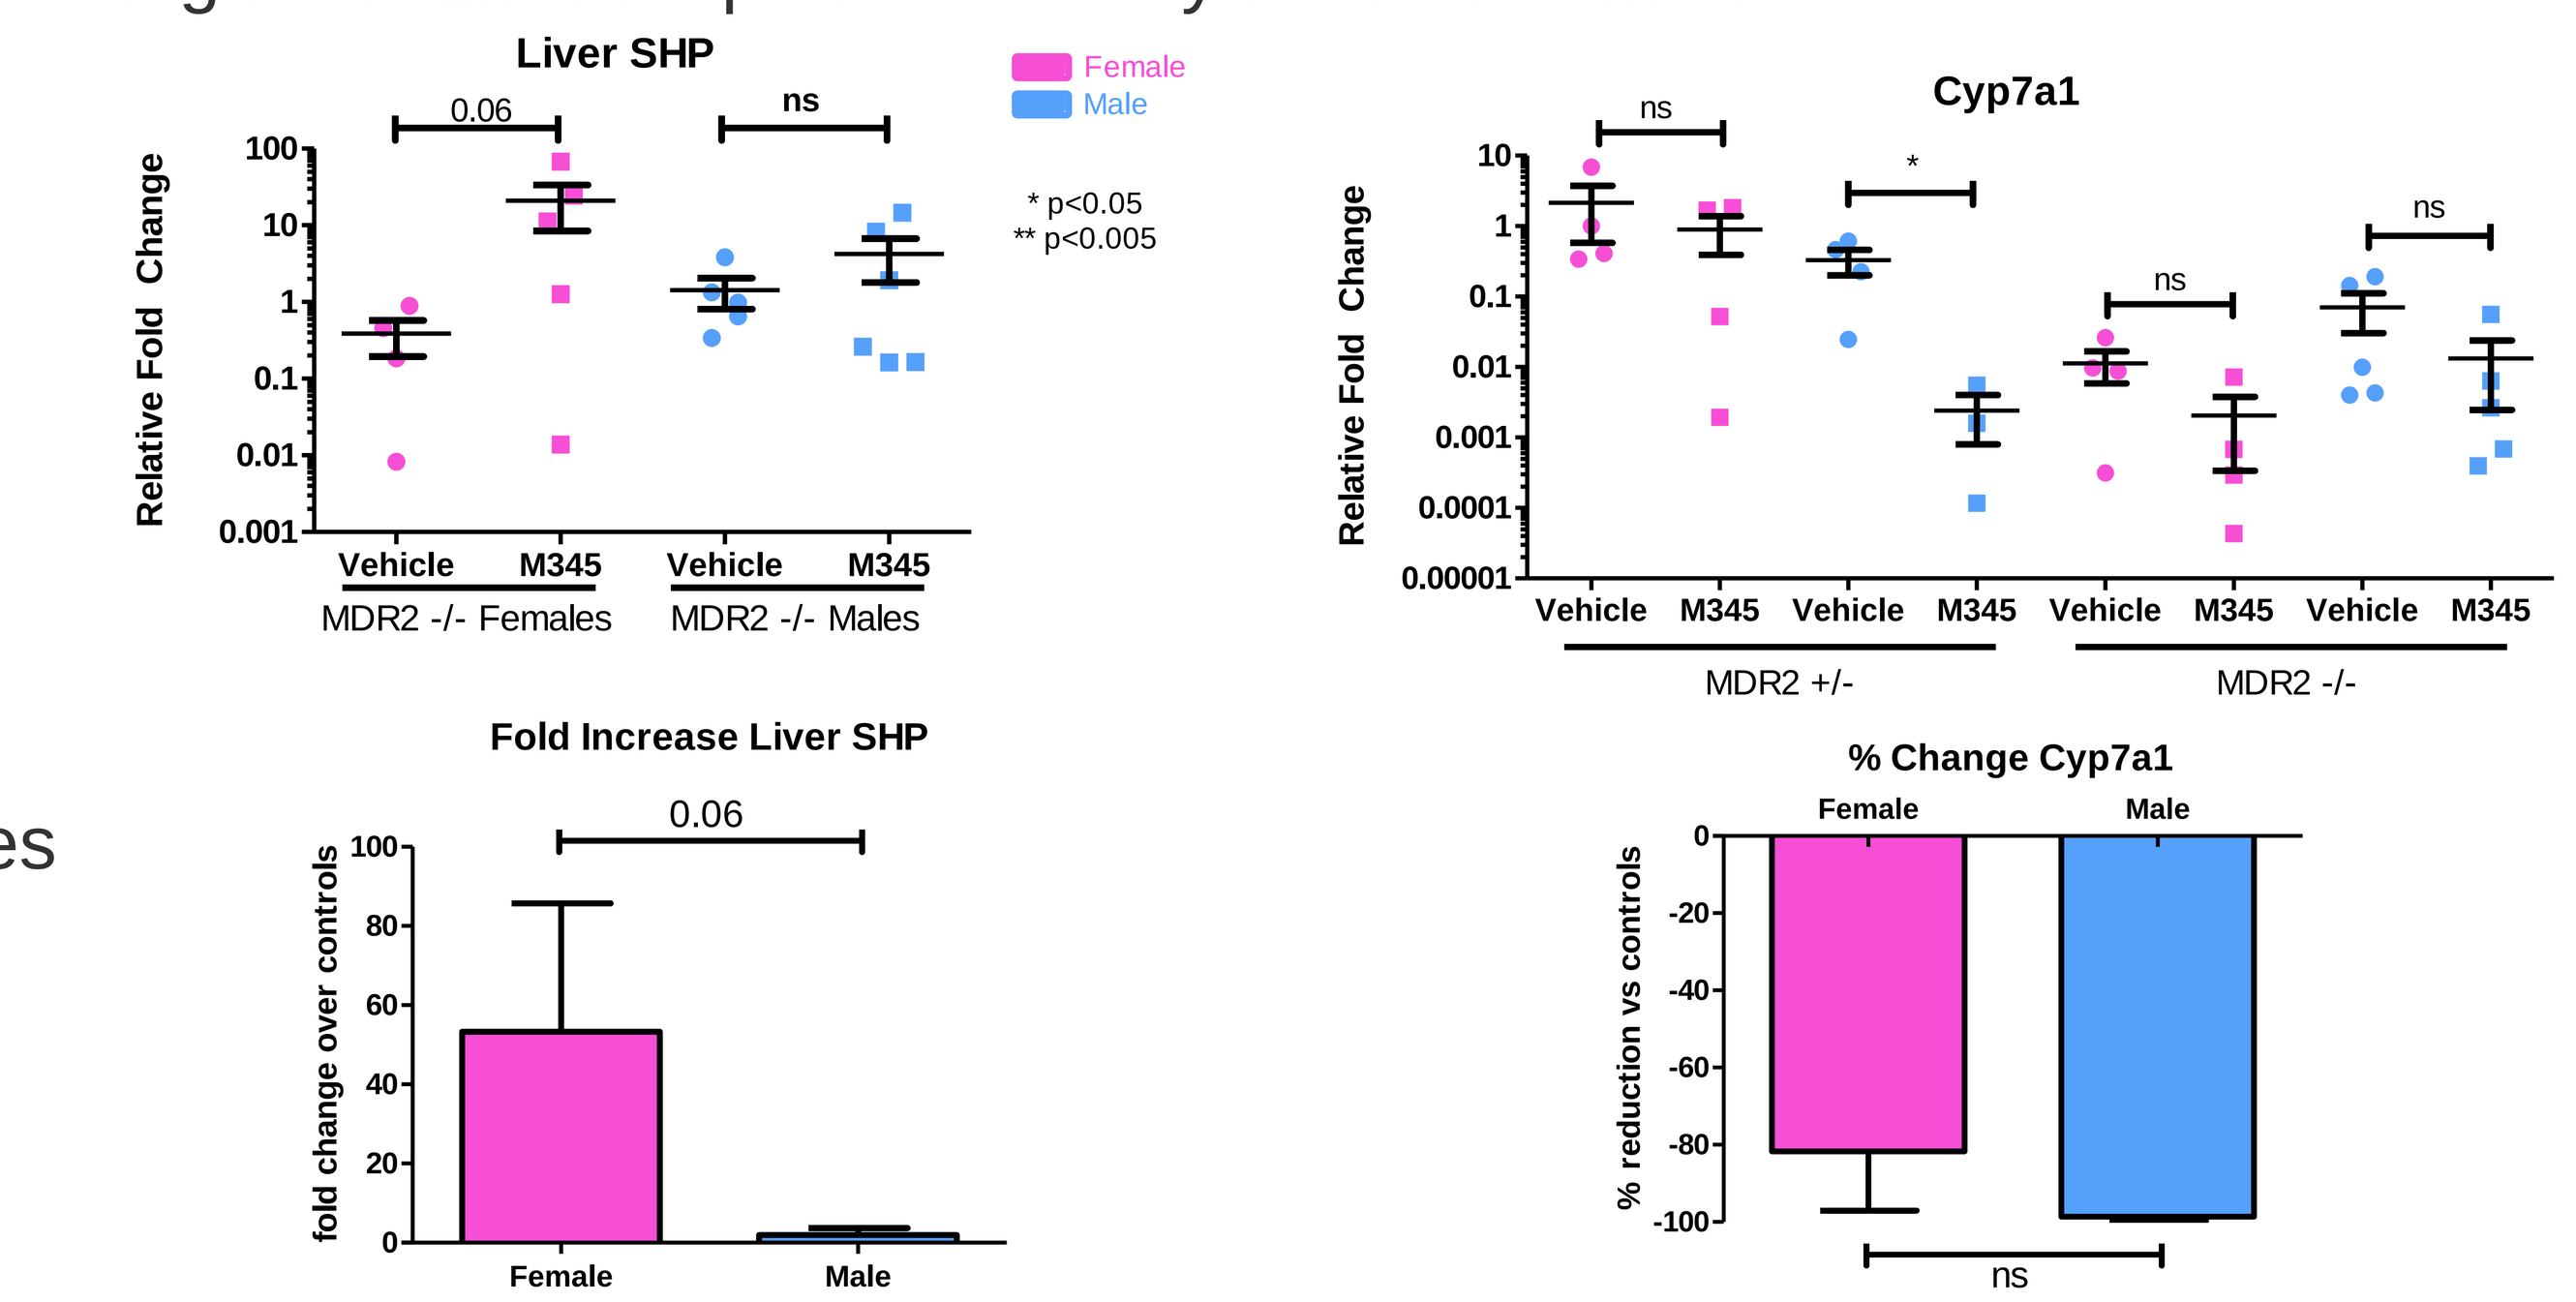
Liver SHP
Cyp7a1
Relative Fold Change
Female
Male
* p<0.05
** p<0.005
0.06
ns
MDR2 -/- Females
MDR2 -/- Males
MDR2 +/-
MDR2 -/-
Fold Increase Liver SHP
fold change over controls
% Change Cyp7a1
% reduction vs controls

Figure 8: Terminal ileum response to 8-day M345 treatment

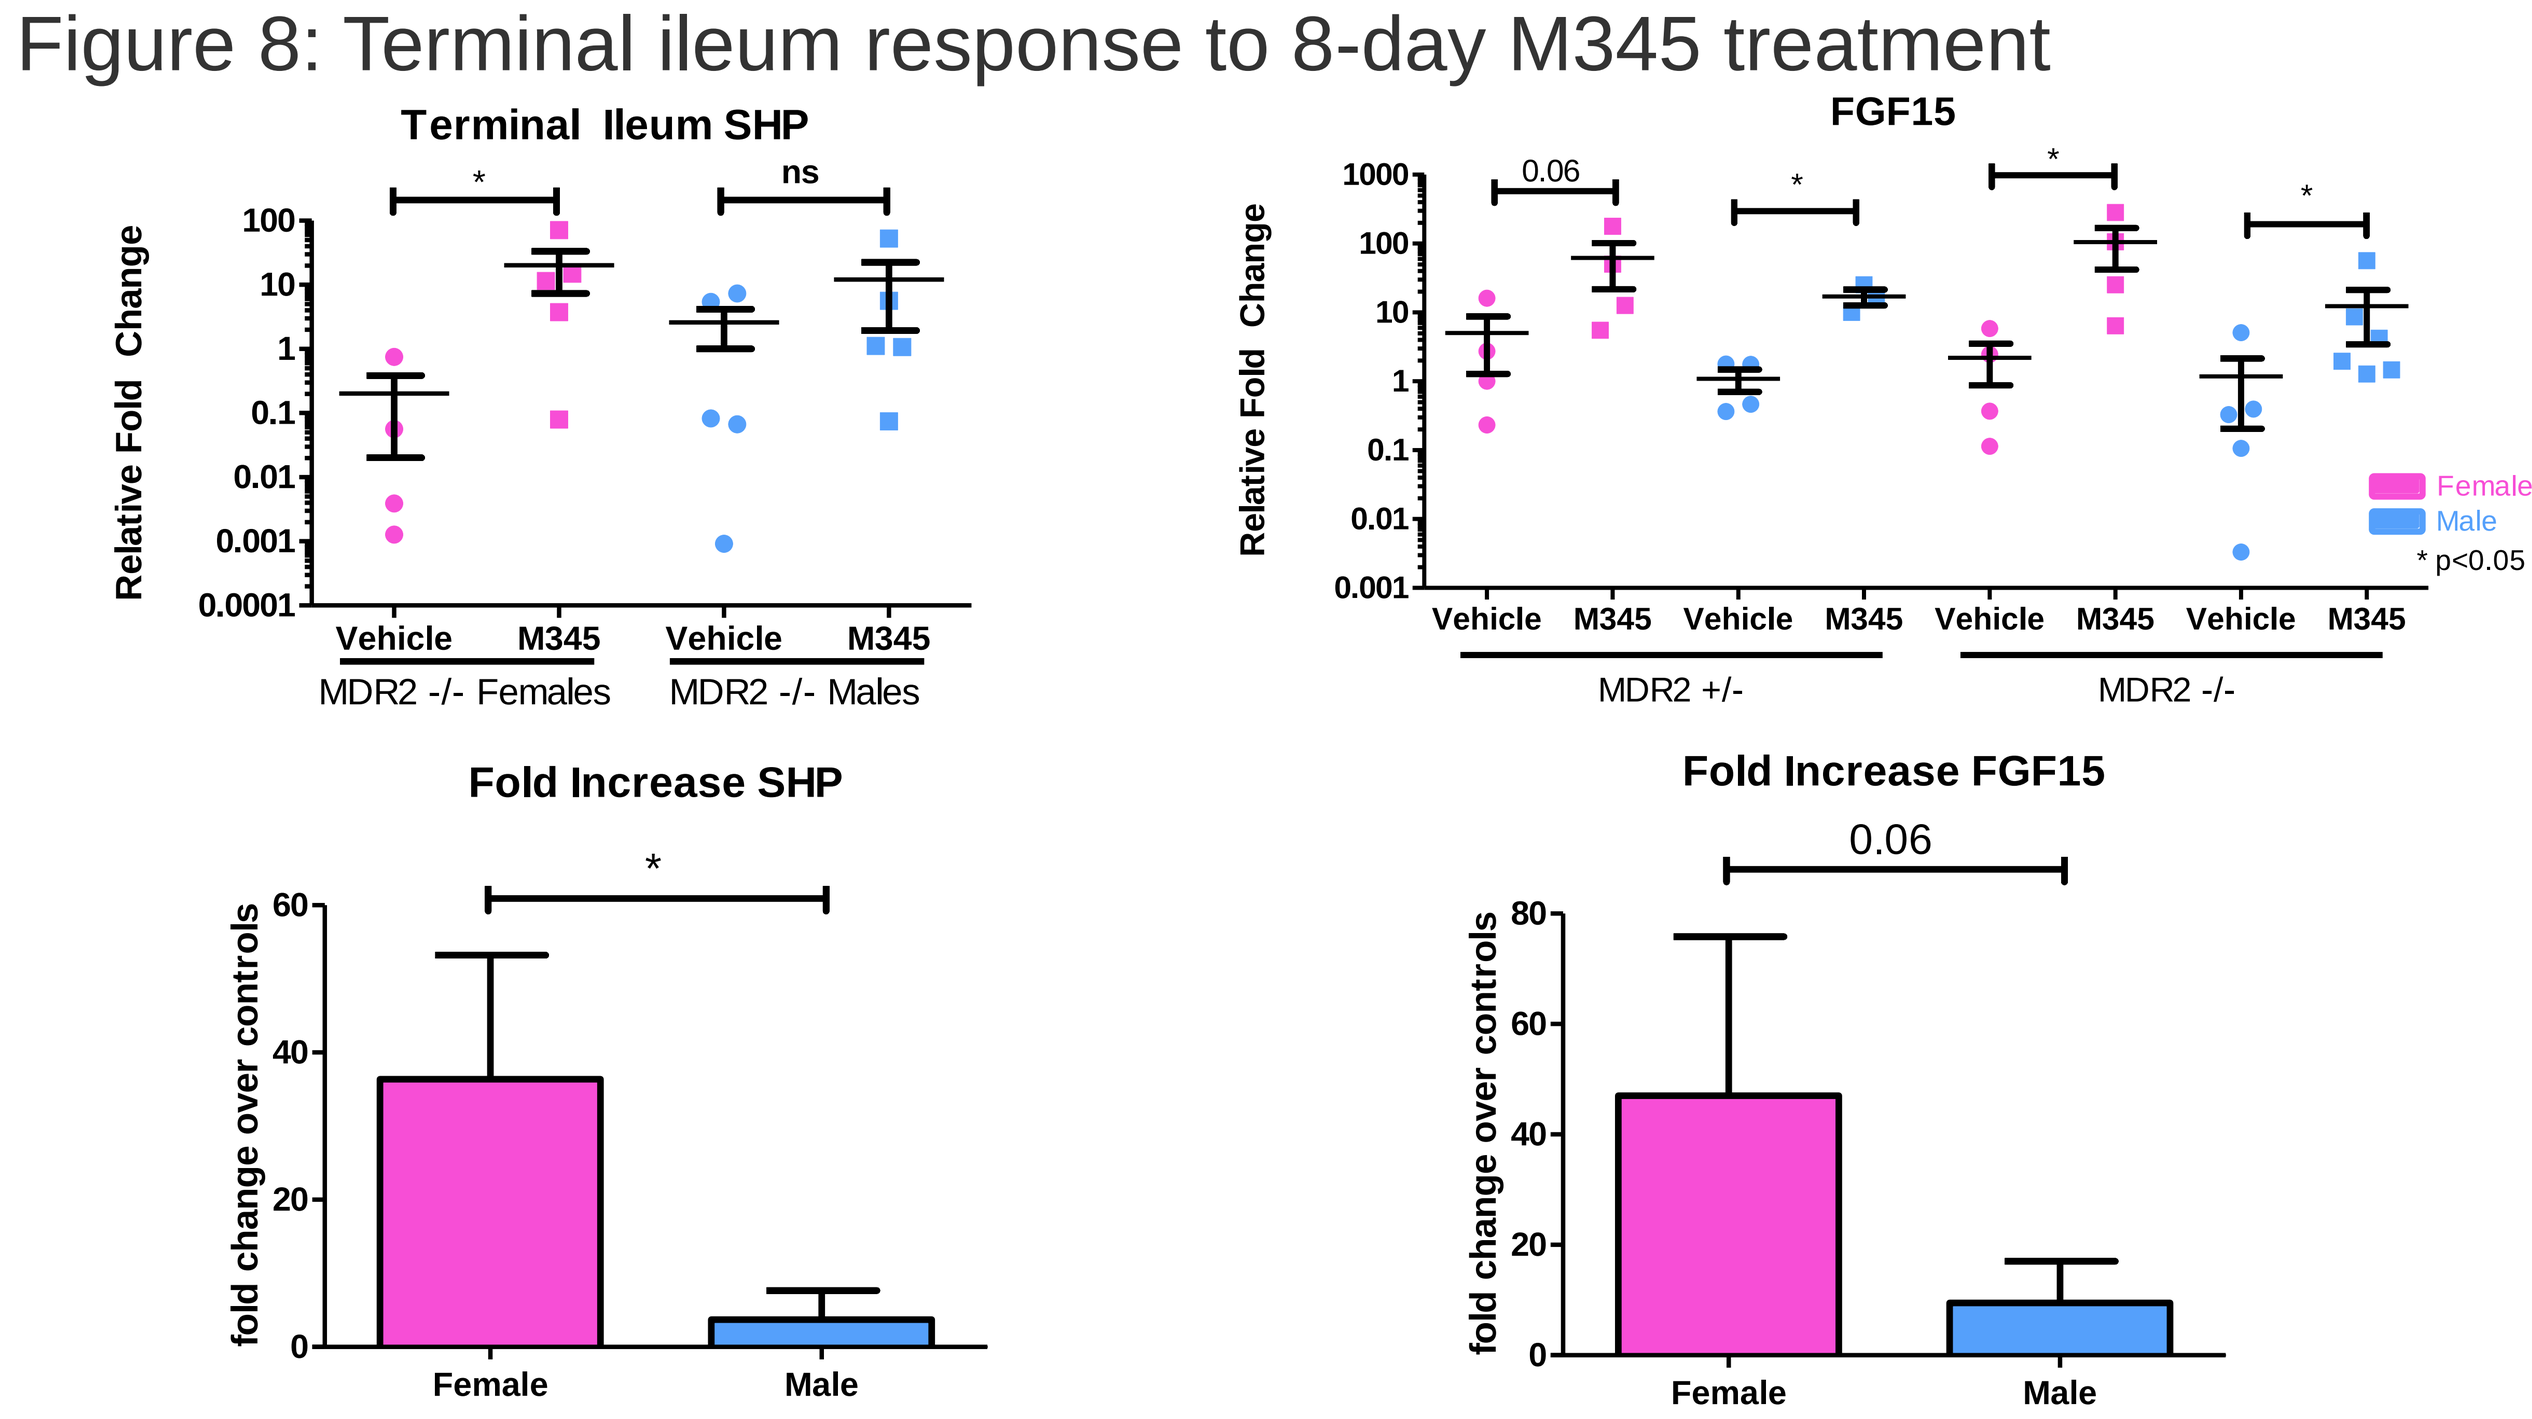
Terminal Ileum SHP
FGF15
Relative Fold Change
Female
Male
* p<0.05
MDR2 -/- Females
MDR2 -/- Males
MDR2 +/-
MDR2 -/-
Fold Increase SHP
Fold Increase FGF15
fold change over controls
0.06

Figure 9: Changes in bile acid and C4 levels in response to M345 treatment

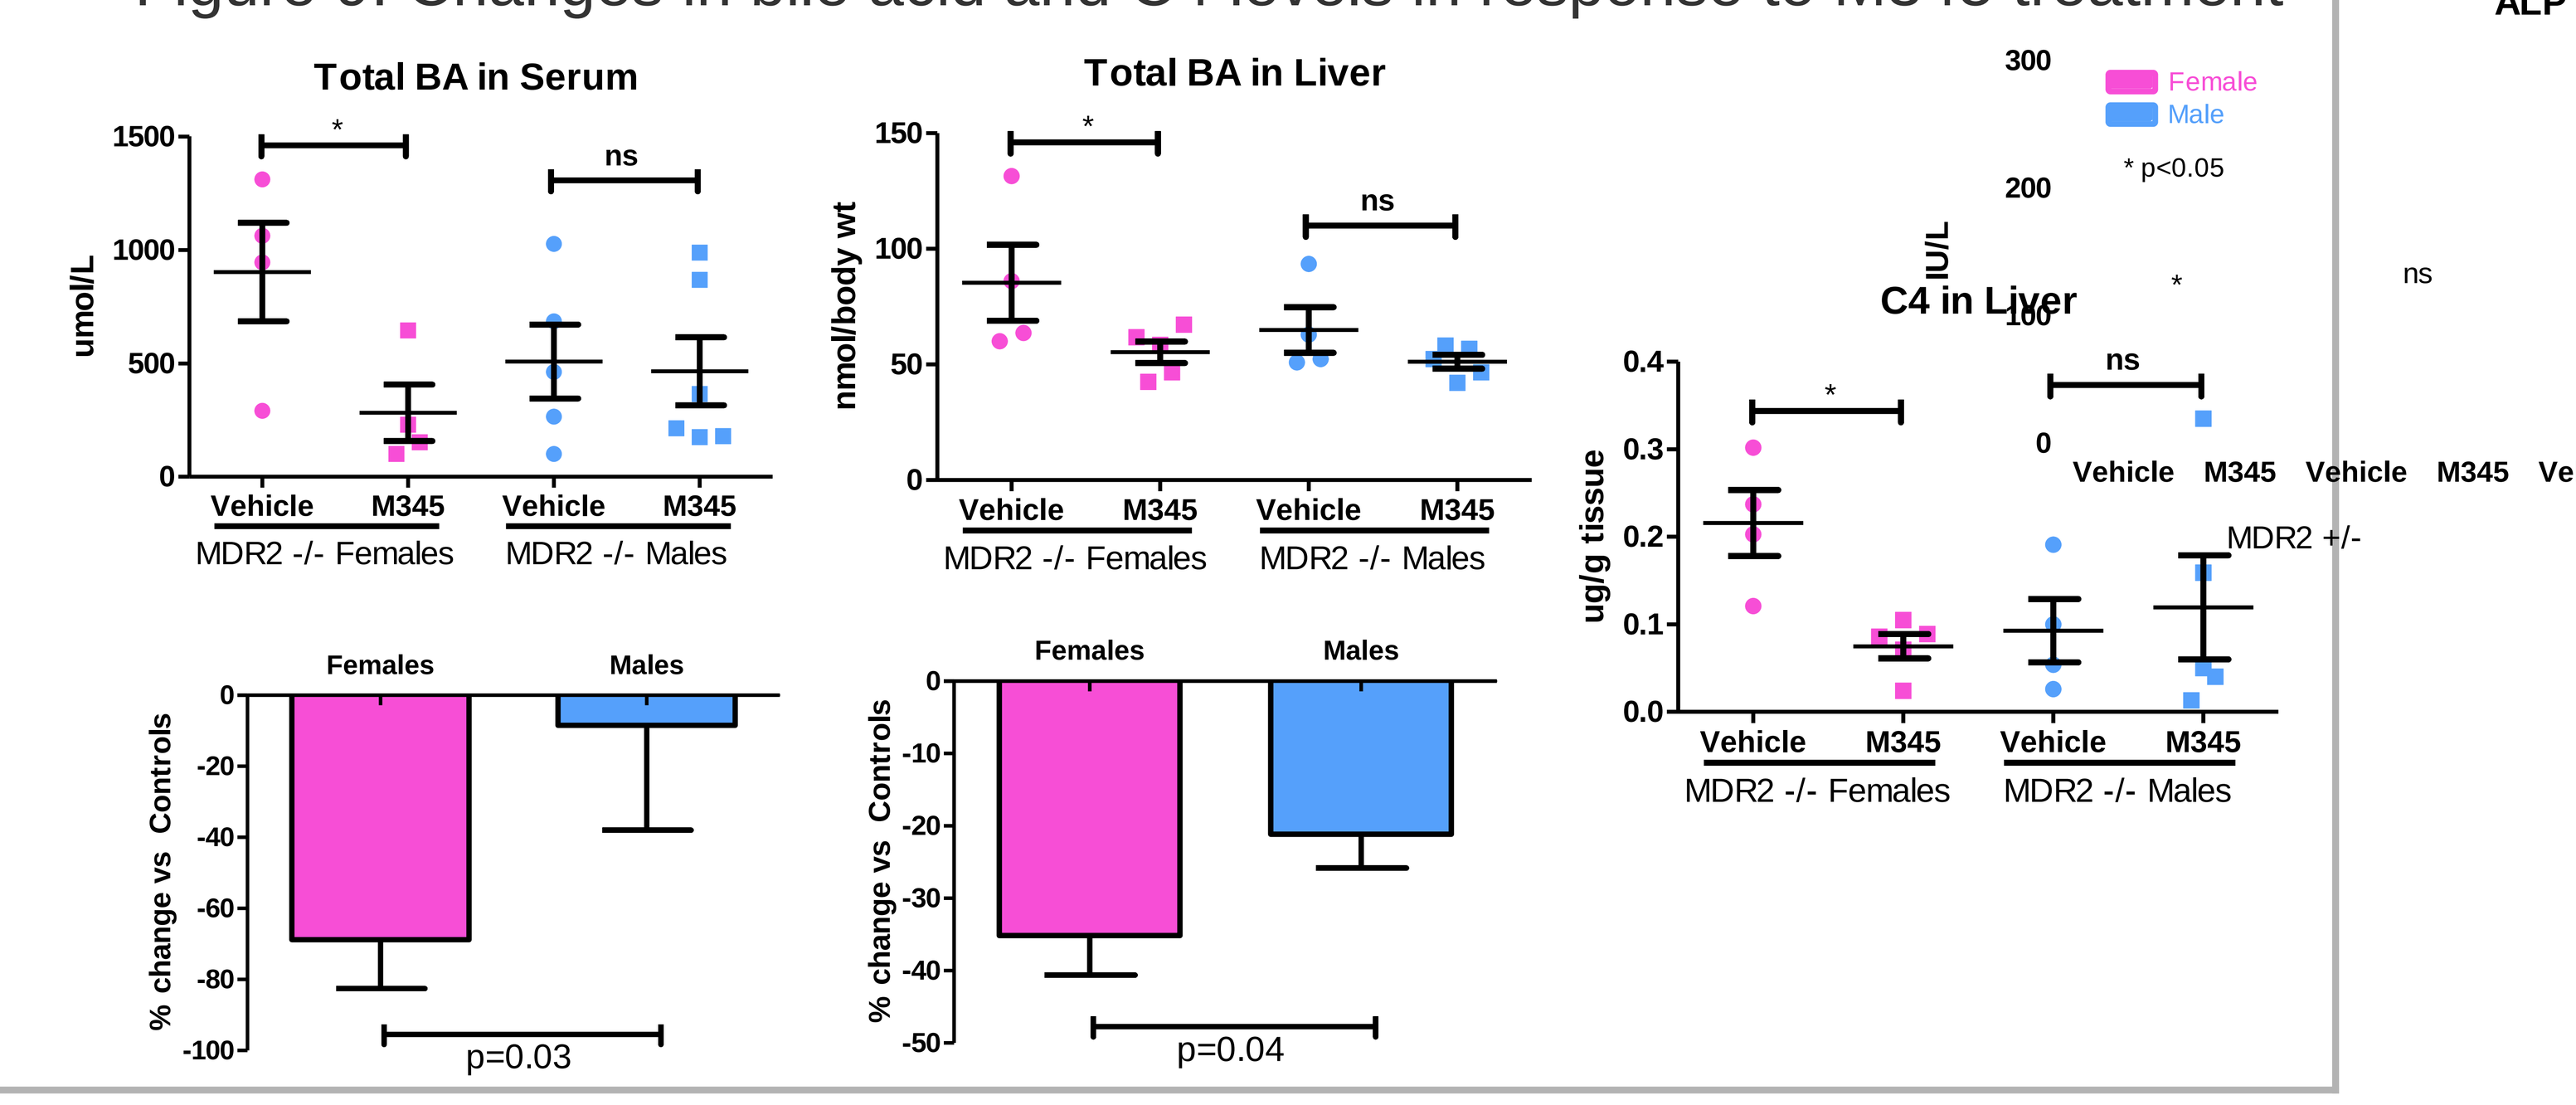
Total BA in Serum
Total BA in Liver
C4 in Liver
umol/L
nmol/body wt
ug/g tissue
MDR2 -/- Females
MDR2 -/- Males
% change vs Controls
Females
Males
p=0.03
p=0.04

Summary

At baseline:
- Female Mdr2-/- display a more severe phenotype of sclerosing cholangitis with higher serum liver biochemistry levels, and aggravated liver bile duct proliferation and fibrosis when compared to age-matched male mice.

Upon short-term M345 treatment, female Mdr2-/- mice:
- Improve weight gain, decrease serum liver biochemistries, and reduced hepatic TNF-α expression
- Induce *Shp* expression and downregulate *Cyp7a1* in the liver, upregulate Shp and *Fgf15* mRNA expression in the terminal ileum, decrease liver C4 as intermediate of de novo bile acid synthesis, which is associated with significant decrease in liver and serum bile acid concentrations

In contrast, upon short-term M345 treatment, male compared with female Mdr2-/- mice:
- Show diminished induction of *Shp* and *Fgf15* in the terminal ileum, and do not reduce their liver or serum BA concentrations
- Fail to gain weight and do not lower significantly serum liver biochemistries despite similar concentrations of M345 in the liver.

Conclusions

Short-term treatment with the FXR agonist M345 decreases liver injury, inflammation, and improves weight gain in female Mdr2-/- mice with severe cholestasis. Attenuated response in male mice may result from diminished FXR activation in the intestine.

Acknowledgements

Project supported by NIH R01DK095001-01 (A.G.M), and the foundation "PSC Partners seeking a cure" (A.G.M). This project was also supported in part by PHS Grant P30 DK078392 (Pathology Research Core) of the Digestive Disease Research Core Center and by the Center for Autoimmune Liver Disease in Cincinnati .