MET409, an Optimized Sustained FXR Agonist, Was Safe and Well-Tolerated in a 14-Day Phase 1 Study in Healthy Subjects

Hubert C. Chen[1], Karl Cremer[1], Eric Bischoff[1], Kyoung-Jin Lee[1], Rozalyn Littler[1], Jeroen van de Wetering[2], Gerhard Arold[2], Minh-Nguyet Duong[2], Ken Song[1]

[1]Metacrine, Inc., San Diego, USA; [2]PRA Health Sciences, Groningen, Netherlands

METACRINE™
3985 Sorrento Valley Blvd., Suite C
San Diego, CA 92121 USA
www.metacrine.com

INTRODUCTION

Farnesoid X receptor (FXR) agonists for the treatment of non-alcoholic steatohepatitis (NASH) have evolved, although each iteration has had drawbacks. FXR agonists have shown increased low-density lipoprotein (LDL) cholesterol levels, as well as moderate to severe pruritus within 14 days of dosing. More recent, non-bile acid agonists are hampered by transient target engagement with once-daily dosing, likely limiting efficacy. Sustained FXR engagement appears key to efficacy based on preclinical and clinical studies. MET409 is an optimized next-generation FXR agonist with features of a novel non-bile acid structure, enhanced potency and sustained FXR engagement.

AIM

We conducted a double-blind, placebo-controlled, single-ascending dose (SAD) and 14-day multiple-ascending dose (MAD) study in healthy male subjects to determine the safety, tolerability, pharmacokinetics (PK) and pharmacodynamics (PD) of MET409.

MATERIAL & METHODS

Healthy men aged 18 to 65 years old were randomized to either MET409 or placebo (6:2 allocation in SAD; 8:2 allocation in MAD). The primary study objective was to assess the safety and tolerability of MET409. The secondary objectives were to establish the PK and PD profiles of MET409, with PD assessed by plasma levels of 7α -hydroxy-4-cholesten-3-one (C4) and fibroblast growth factor 19 (FGF19).

RESULTS

Both single (20-400 mg) and multiple (20-150 mg) doses of MET409 were safe and well tolerated. There were no serious adverse events, and the severity and frequency of adverse events were comparable between MET409 and placebo. Mild pruritus, observed only at 100 and 150 mg levels, did not require medical intervention or dosing interruption.

MET409 exhibited sustained PK and PD profiles with once-daily dosing. There were dose-dependent increases in maximum concentration (C_{max}) and exposure (AUC_{0-24}). Time-dependent increase in exposure was observed, with steady-state reached after ~10-14 days. Elimination half-life ($T_{1/2}$) was ~15 hrs on Day 1 and ≥50-100 hrs on Day 14 at expected therapeutic levels (50-100 mg).

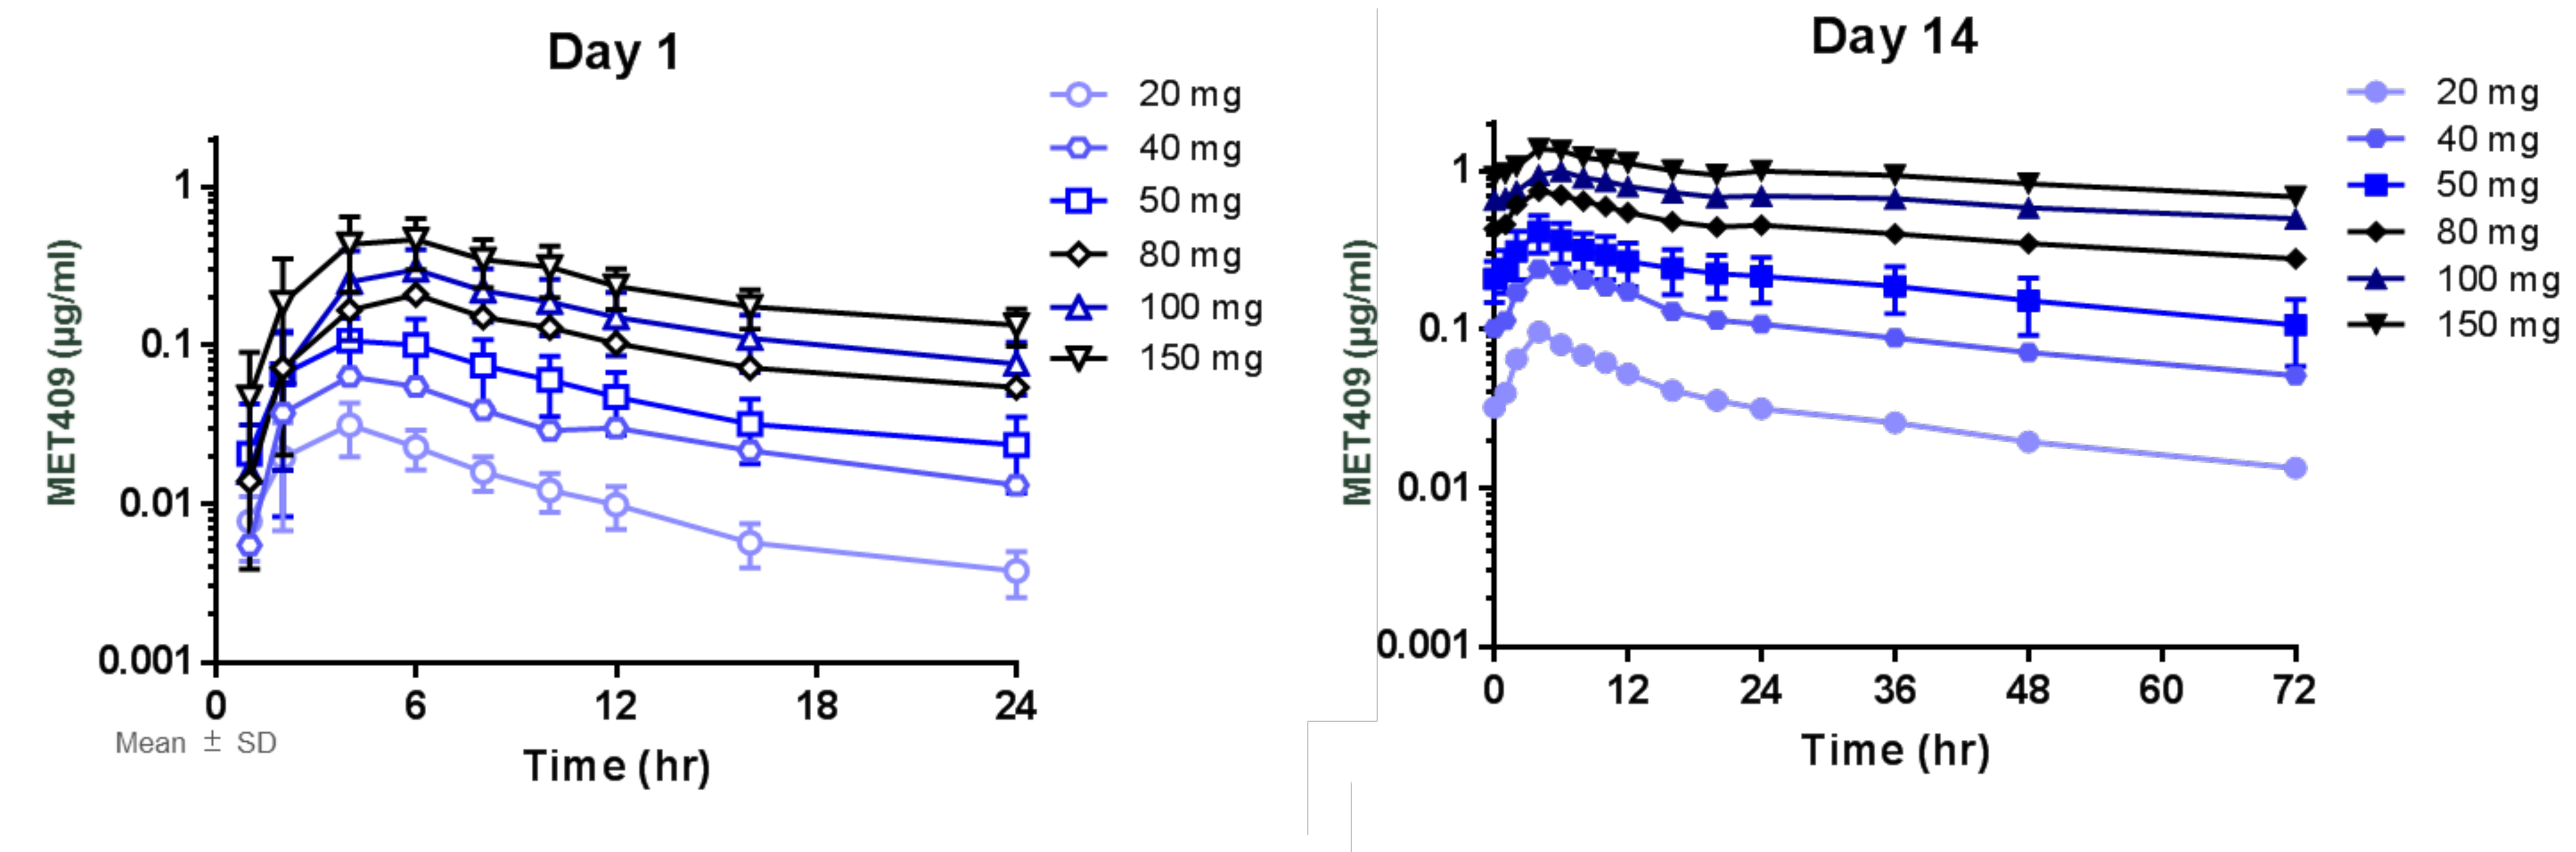

Day 1
Day 14
MET409 (µg/ml)
Time (hr)
Mean ± SD
20 mg
40 mg
50 mg
80 mg
100 mg
150 mg

MET409, at ≥50 mg, suppressed plasma C4 levels (↓~80-99%) relative to placebo after 14 days of daily dosing, with suppression observed throughout 24 hrs. Increased FGF19 levels were also observed with MET409.

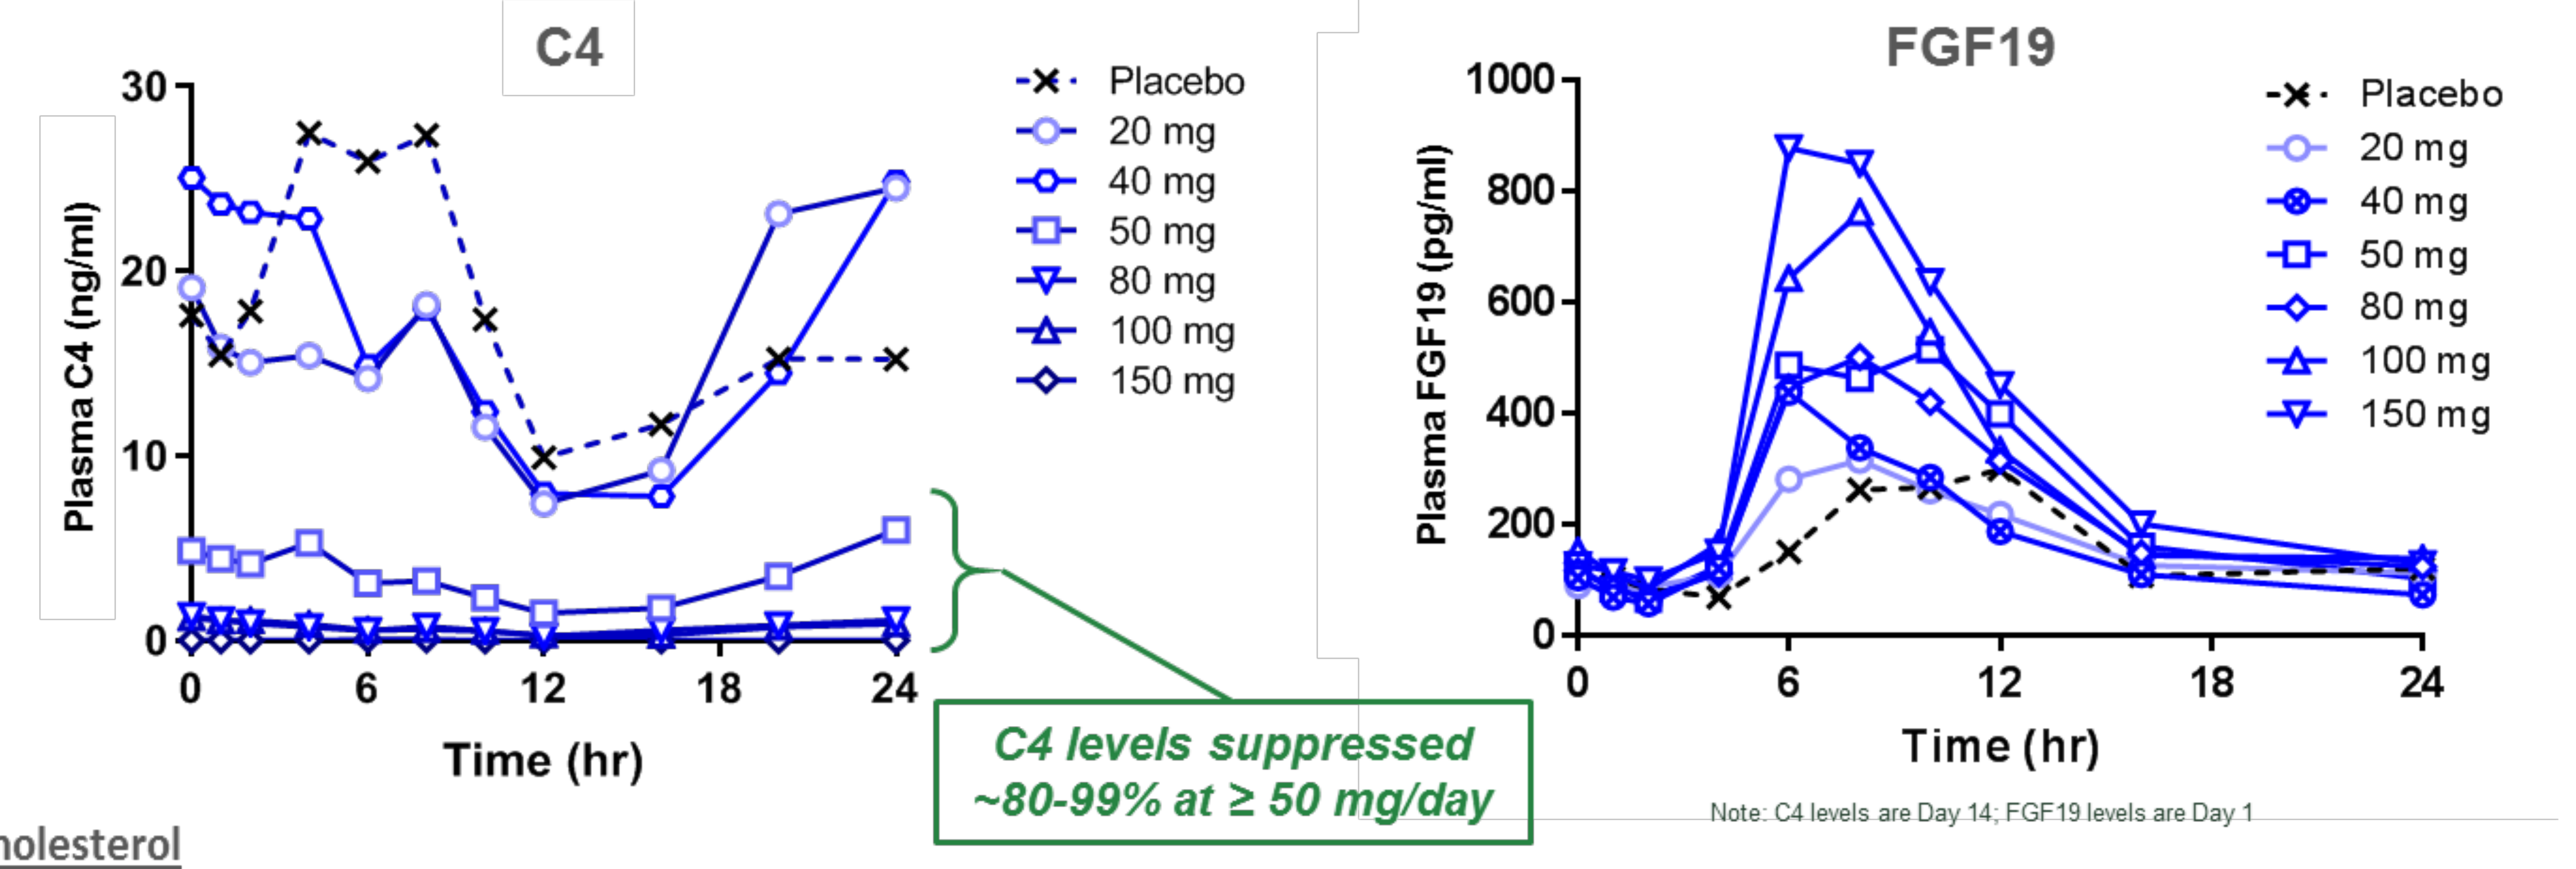

C4
Plasma C4 (ng/ml)
Time (hr)
Placebo
20 mg
40 mg
50 mg
80 mg
100 mg
150 mg
C4 levels suppressed ~80-99% at ≥ 50 mg/day
FGF19
Plasma FGF19 (pg/ml)
Note: C4 levels are Day 14; FGF19 levels are Day 1

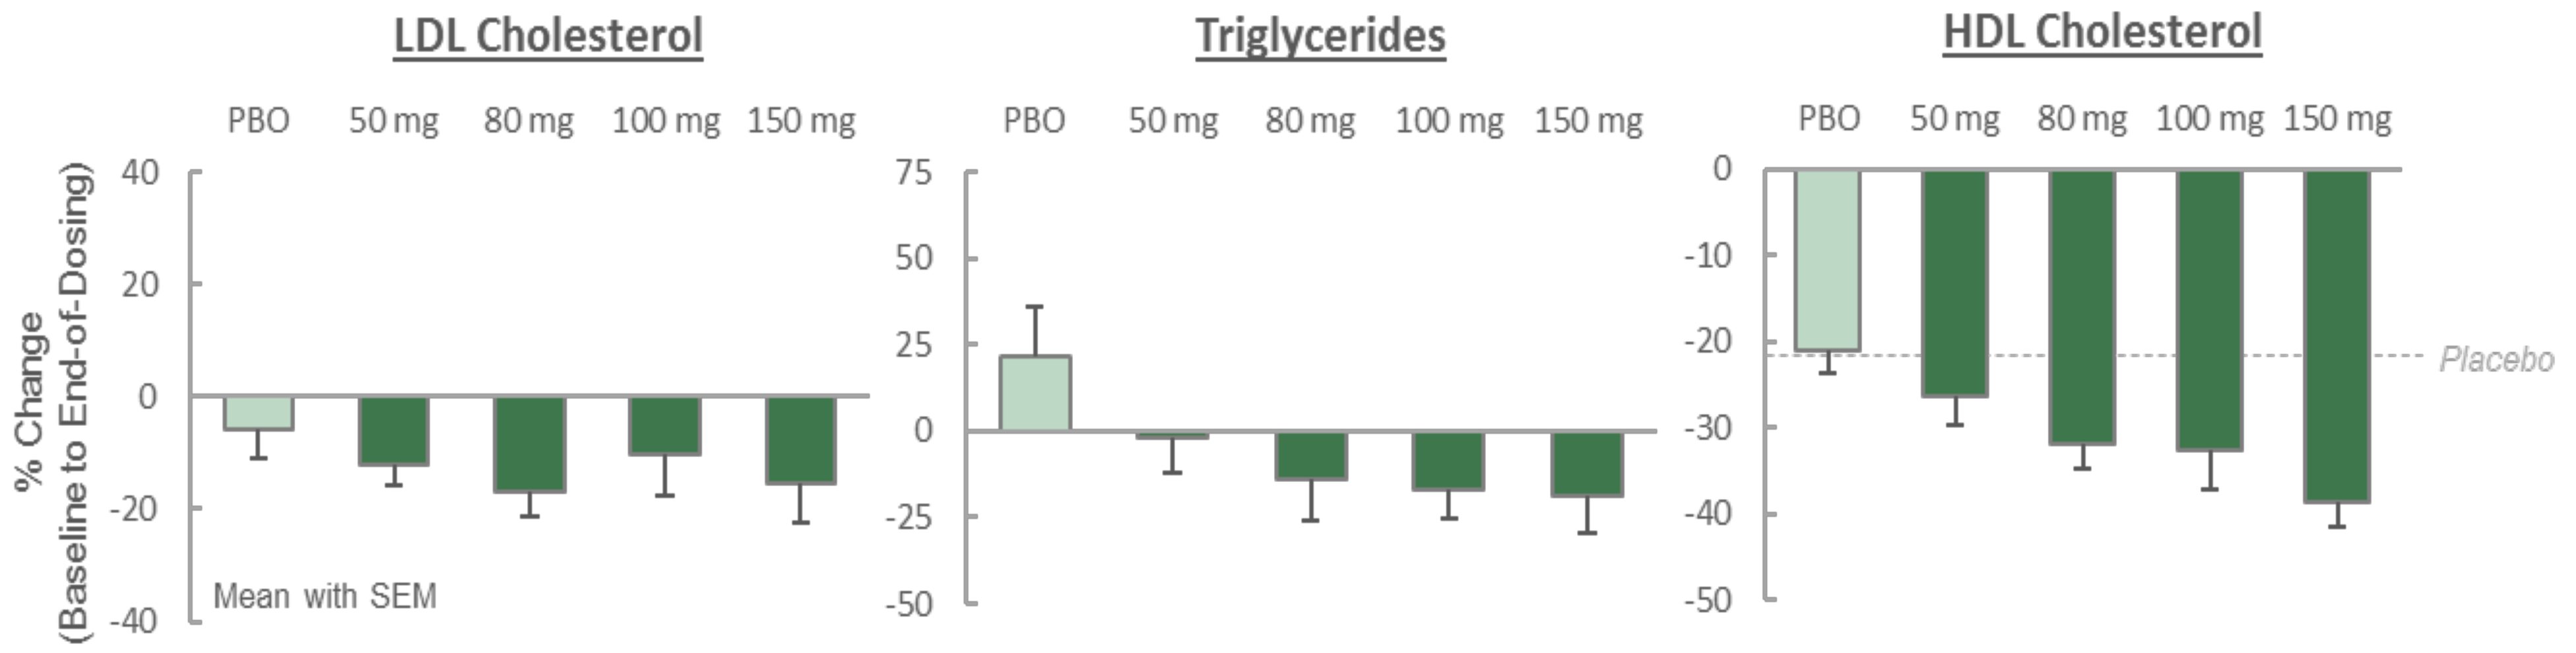

LDL Cholesterol
Triglycerides
HDL Cholesterol
PBO
50 mg
80 mg
100 mg
150 mg
% Change (Baseline to End-of-Dosing)
Mean with SEM
Placebo

MET409 did not increase LDL cholesterol at pharmacologically active dose levels, a differentiating attribute relative to other FXR agonists and FGF19 analogs. There were mild, dose-dependent decreases in high-density lipoprotein (HDL) cholesterol and triglycerides.

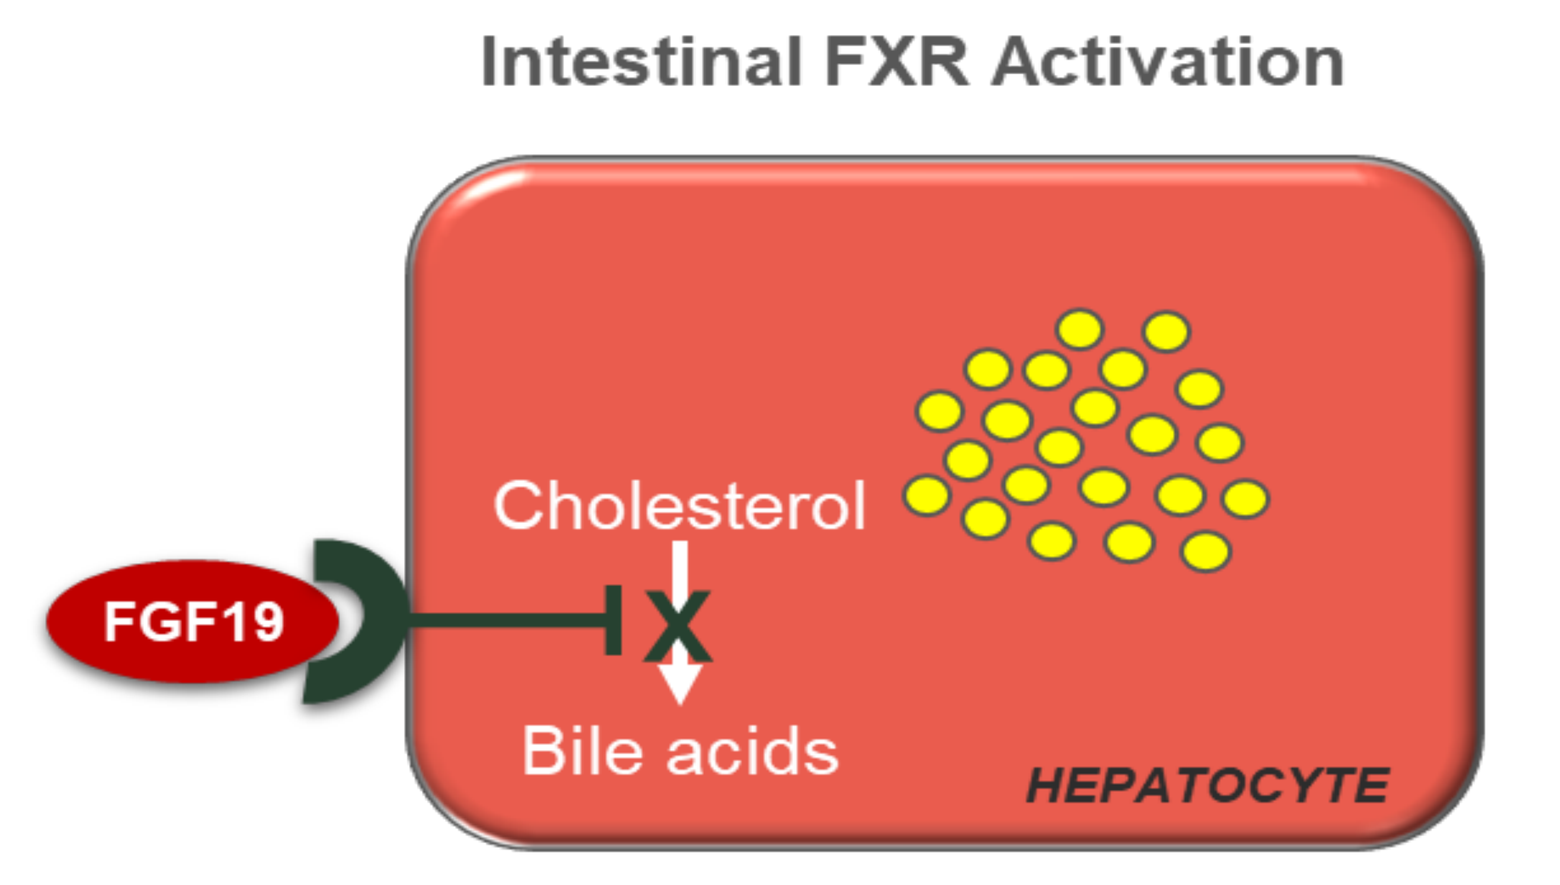

Intestinal FXR Activation
FGF19
Cholesterol
Bile acids
HEPATOCYTE

Intestinal FXR activation releases FGF19 which binds receptors on hepatocytes → suppression of bile acid synthesis → increase LDL cholesterol

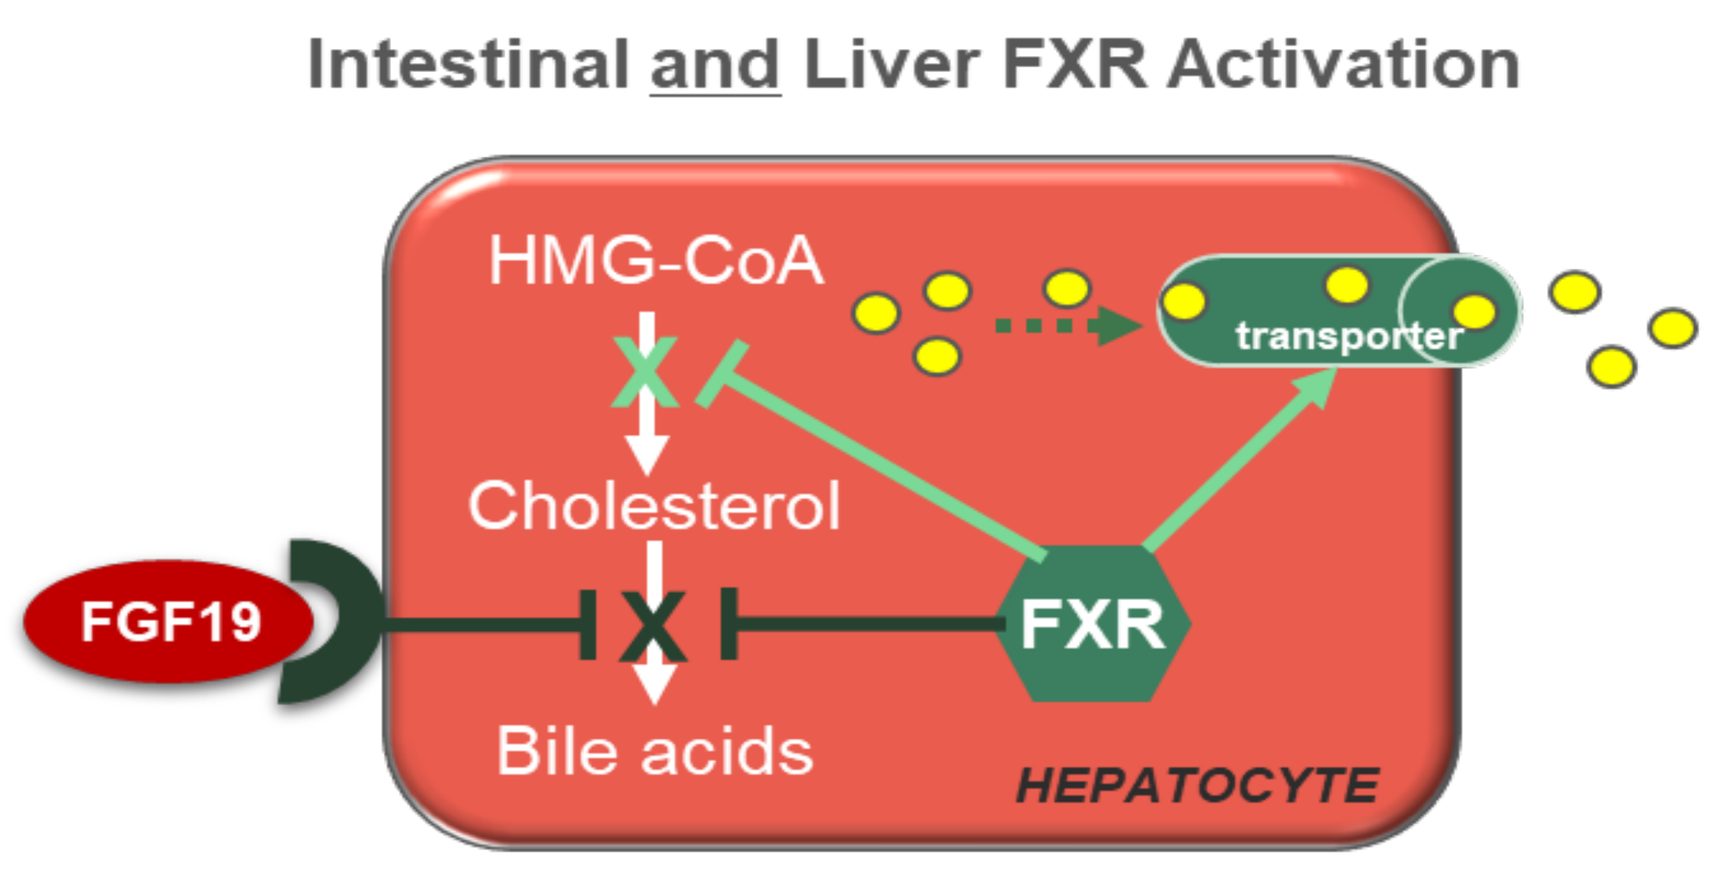

Intestinal and Liver FXR Activation
HMG-CoA
transporter
Cholesterol
FGF19
FXR
Bile acids
HEPATOCYTE

Liver FXR activation counters FGF19-mediated increase in LDL cholesterol, possibly due to:

- *Less production via HMG-CoA reductase repression*
- *More excretion into bile via transporters*
- *More clearance via PCSK9 repression (not shown)*

MET409's lack of adverse effects on LDL cholesterol may be due its optimized, sustained activity on liver FXR. In contrast to other FXR agonists that primarily activate intestinal FXR or have transient liver FXR engagement (left figure), MET409's sustained liver FXR activity counters FGF19-mediated increase in LDL cholesterol (right figure).

CONCLUSION

MET409, an optimized FXR agonist, demonstrated sustained FXR engagement and an encouraging safety and tolerability profile – including a lack of adverse impact on LDL cholesterol levels – in healthy subjects. These results support a thesis that sustained FXR agonism delivers a differentiated, best-in-class profile for the treatment of NASH.