FXR AGONIST M480, EXHIBITS EFFICACY IN MULTIPLE MODELS OF INFLAMMATORY BOWEL DISEASE (IBD) BY INCREASING EXPRESSION OF ANTI-MICROBIAL GENES AND REDUCING INFLAMMATORY CYTOKINE EXPRESSION IN THE GUT

Xueqing Liu[1], Harry Dedman[1], Robert O'Connell[2], Alison Bendele[2], Connor Ohlsen[2], Joshua D. Boyer[3], Steve Govek[1], Johnny Nagasawa[1], Karensa Douglas[1], Andiliy Lai[1], Angelica Milik[1], Nhin Lu[1], Jing Qian[1], Alvaro Ortiz[1], Douglas Zook[1] , Kyoung-Jin Lee[1], Nicholas Smith[1], Hubert Chen[1], Brandee Wagner[1], Ken Song[1]

1. Metacrine, San Diego, CA; 2. Bolder BioPath, Inc., Boulder, CO.; 3. University of California San Diego

3985 Sorrento Valley Blvd., Suite C
San Diego, CA 92121
www.metacrine.com

INTRODUCTION

The farnesoid X receptor (FXR) is a ligand-activated nuclear hormone receptor that is highly expressed in the gastrointestinal tract. FXR agonists have shown efficacy in acute DSS and TNBS chemically induced IBD models. We have previously demonstrated efficacy with M480, a potent, novel FXR agonist, in the adoptive transfer (AT) model of colitis.

AIMS

The aims of this study were to further examine the efficacy and mechanism of M480 in multiple chronic IBD models (i.e. AT colitis, Mdr1a$^{-/-}$ colitis, and SAMP1/YitFc ileitis).

MATERIAL & METHODS

AT colitis was induced by transferring CD4+CD45RB^{hi} T-cells to C.B-17 SCID mice. Mdr1a$^{-/-}$ and SAMP1/YitFc mice exhibited spontaneous colitis and ileitis, respectively. All studies were run in therapeutic treatment mode with M480 and positive controls anti-IL-12p40 or prednisolone started at 21 days post AT, or beginning when disease is present at week 8-9 in Mdr1a$^{-/-}$ mice, and week 10 in SAMP1/YitFc mice. M480 (10 mg/kg) or prednisolone (1mg/kg) were orally dosed daily (n=8-11). Anti-IL-12p40 (0.5mg/mouse) was dosed intraperitoneally weekly (n=5). All treatments lasted for 4 weeks. Effects on colitis were assessed by terminal colon weight to length ratio (W/L) and colon histopathology scoring. Efficacy on ileitis was assessed by ileum histopathology scoring.

CONCLUSIONS

M480 reversed loss of body weight and significantly improved colon histopathology in AT and Mdr1a$^{-/-}$ colitis models. Gene expression analysis revealed a significant decrease in cytokine IL-6 mRNA in colon, correlating with decreased inflammatory cell infiltration. FXR activation with M480 led to increased expression of anti-microbial genes Nos2 and Ang in multiple models. In the SAMP1/YitFc ileitis model, M480 exhibited a trend of improved ileitis.

FXR agonist M480 exhibits efficacy in multiple chronic IBD models. Mechanistic analysis suggests FXR activation increases anti-microbial activity, creates a chemical barrier that protects the host against invading microbes and decreases inflammation. These findings support the development of FXR agonists as novel oral therapeutics for treating IBD.

RESULTS

Efficacy of M480 in Adoptive Transfer Model

Change in Body Weights from Baseline (Day 0)

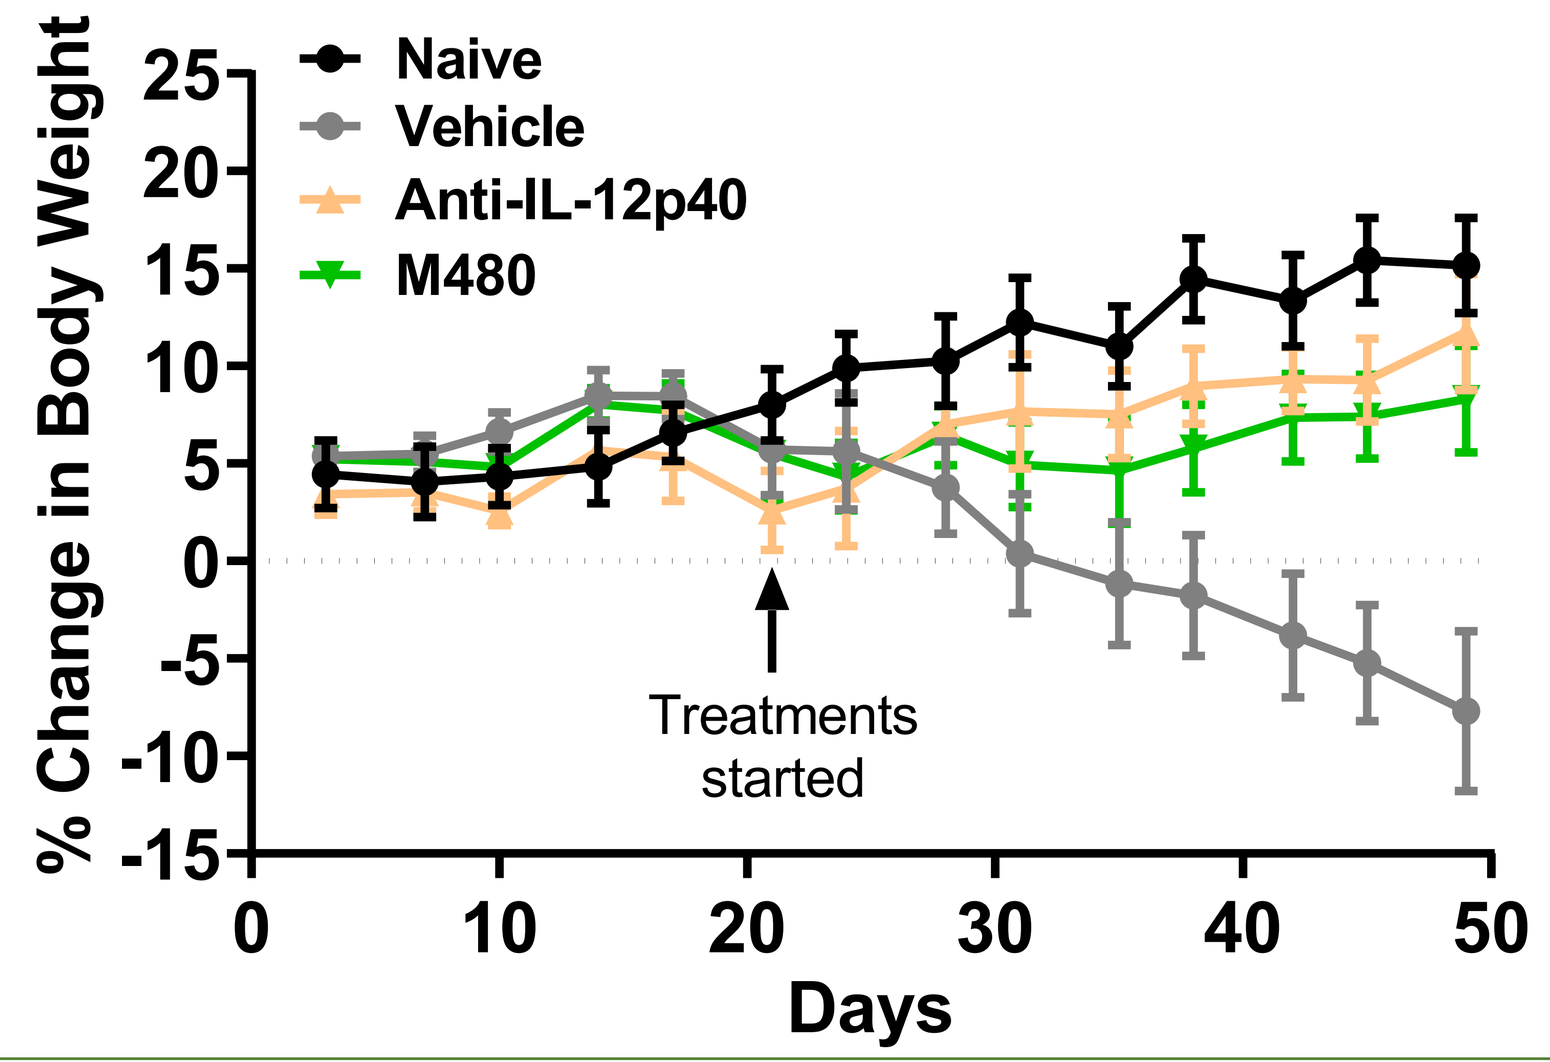

% Change in Body Weight
Naive
Vehicle
Anti-IL-12p40
M480
Treatments started
Days

Improved Colon Histopathology By M480

Treatments	Colon W/L (mg/cm)	Inflammation Score (0-5)	Hyperplasia Score (0-5)	Gland loss Score (0-5)	Histopathology Sum (0-20)
Naïve	19.6	0.0	0.0	0.0	0.0
Vehicle	68.1[a]	4.6[a]	4.0[a]	2.2[a]	11.0[a]
Anti-IL-12p40	31.5[b]	1.5[b]	0.4[b]	0.5[b]	2.4[b]
M480	37.3[b]	1.8[b]	1.2[b]	0.6[b]	3.5[b]

Data shown is mean; a, p<0.05, vs Naïve, b, p<0.05, vs Vehicle, One-Way ANOVA

Efficacy of M480 in Mdr1a$^{-/-}$ Model

Disease Activity Index (DAI *) Time Course

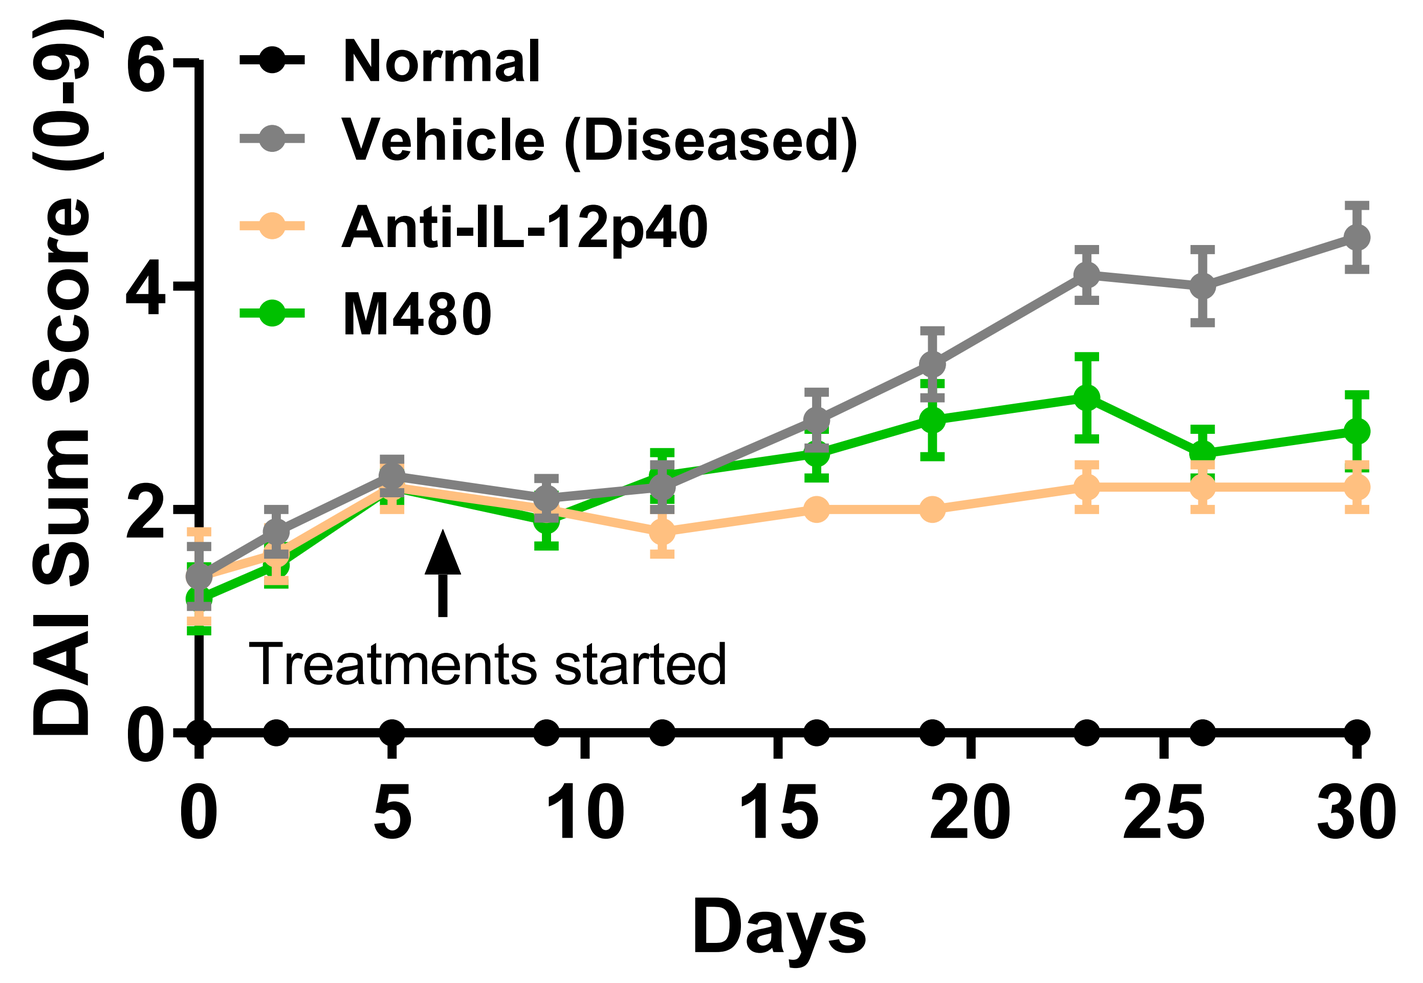

DAI Sum Score (0-9)
Normal
Vehicle (Diseased)
Anti-IL-12p40
M480
Treatments started
Days

*** DAI Sum includes:**
- **Prolapse: 0-3**
- **Stool Consistency: 0-3**
- **Occult Blood: 0-3**

Change in Body Weights From Baseline (Day 0)

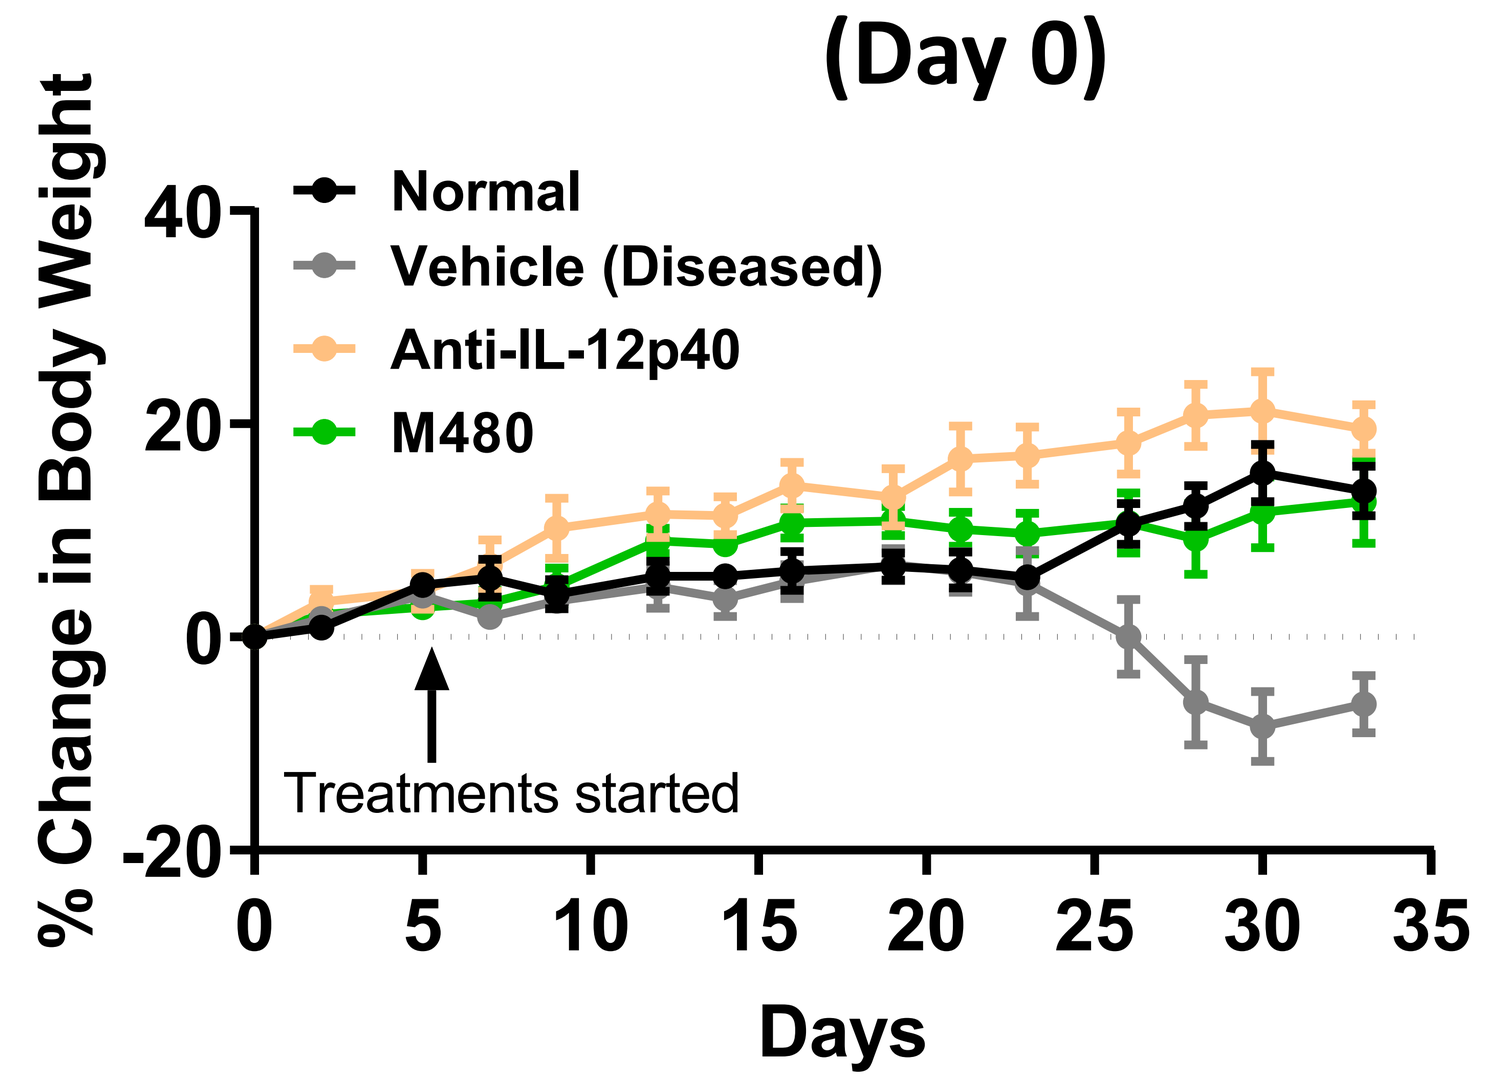

% Change in Body Weight
Normal
Vehicle (Diseased)
Anti-IL-12p40
M480
Treatments started
Days

Improved Colon Histopathology By M480

Treatments	Colon W/L (mg/cm)	Inflammation Score (0-5)	Hyperplasia Score (0-5)	Gland loss Score (0-5)	Histopathology Sum (0-20)
Naïve	23.2	0	0	0	0
Vehicle	96.1[a]	4.8[a]	3.6[a]	2.6[a]	11.9[a]
Anti-IL-12p40	36.6[b]	1.2[b]	0.5[b]	0.3[b]	1.9[b]
M480	62.1[b]	3.5[b]	2.2[b]	1.3[b]	7.1[b]

Data shown is mean; a, p<0.05, vs Naïve, b, p<0.05, vs Vehicle, One-Way ANOVA

Efficacy of M480 in SAMP1/YitFc Model

SAMP1/YitFc Model Ileum Histopathology

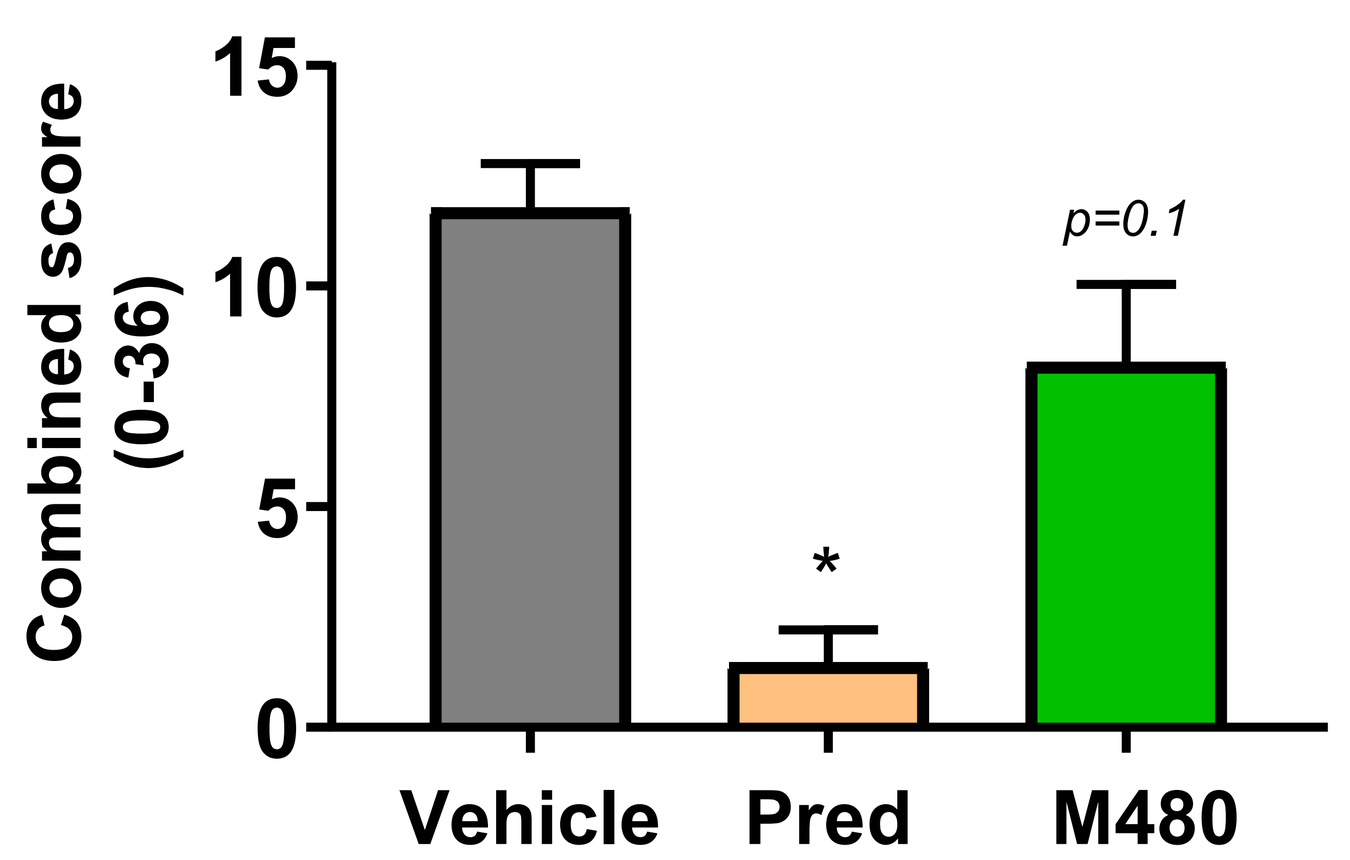

Combined score (0-36)
p=0.1
Vehicle
Pred
M480

** p<0.05 vs Vehicle*

Trend of Improved Ileum Histopathology By M480

Treatments	Active Inflammation Score (0-12)	Chronic Inflammation Score (0-12)	Villus Distortion Score (0-12)	Histopathology Sum (0-36)
Vehicle	4.75	1.0	6.0	11.8
Prednisolone	0.9[a]	0.2[a]	0.4[a]	1.5[a]
M480	3.3	0.6	4.4	8.3

Data is mean; a, p<0.05, vs Vehicle, One-Way ANOVA

Improved Histopathology and Reduced Inflammation in Multiple Models

Histopathology Summary

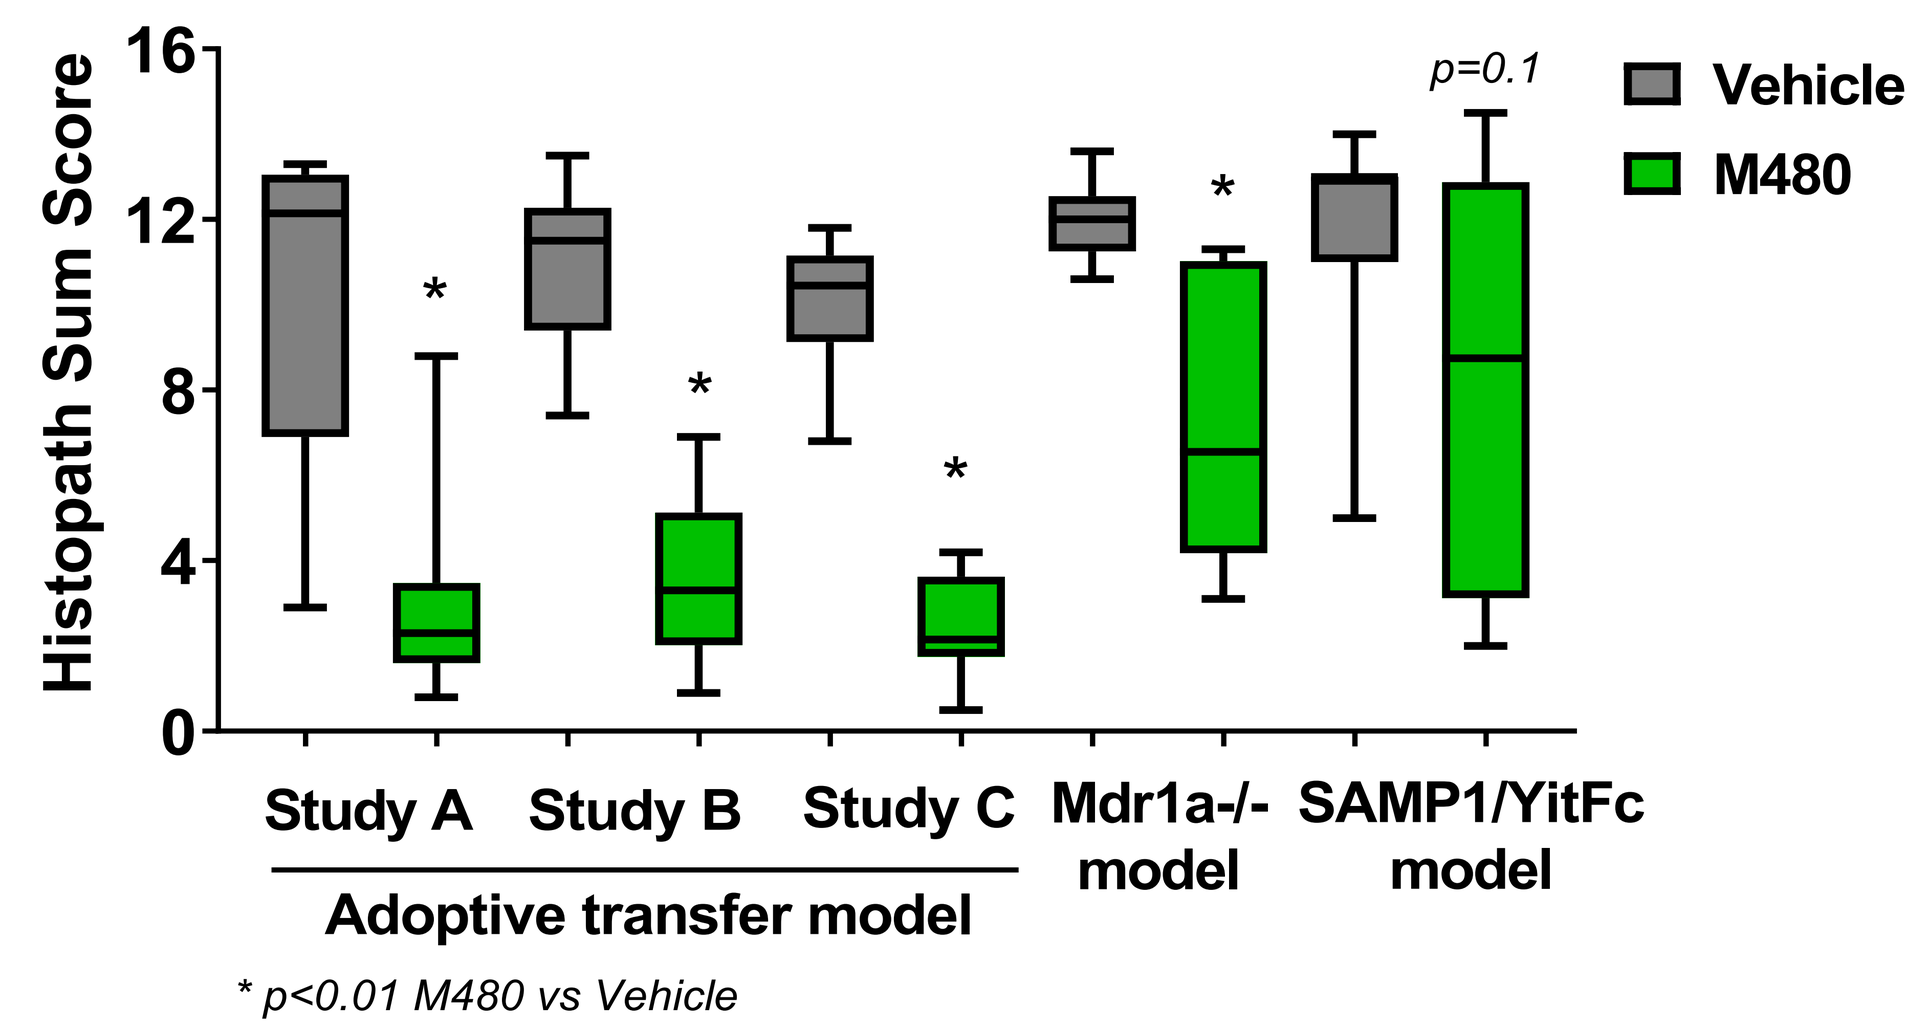

Histopath Sum Score
p=0.1
Vehicle
M480
Study A
Study B
Study C
Mdr1a-/- model
SAMP1/YitFc model
Adoptive transfer model

** p<0.01 M480 vs Vehicle*

IL-6 mRNA Expression

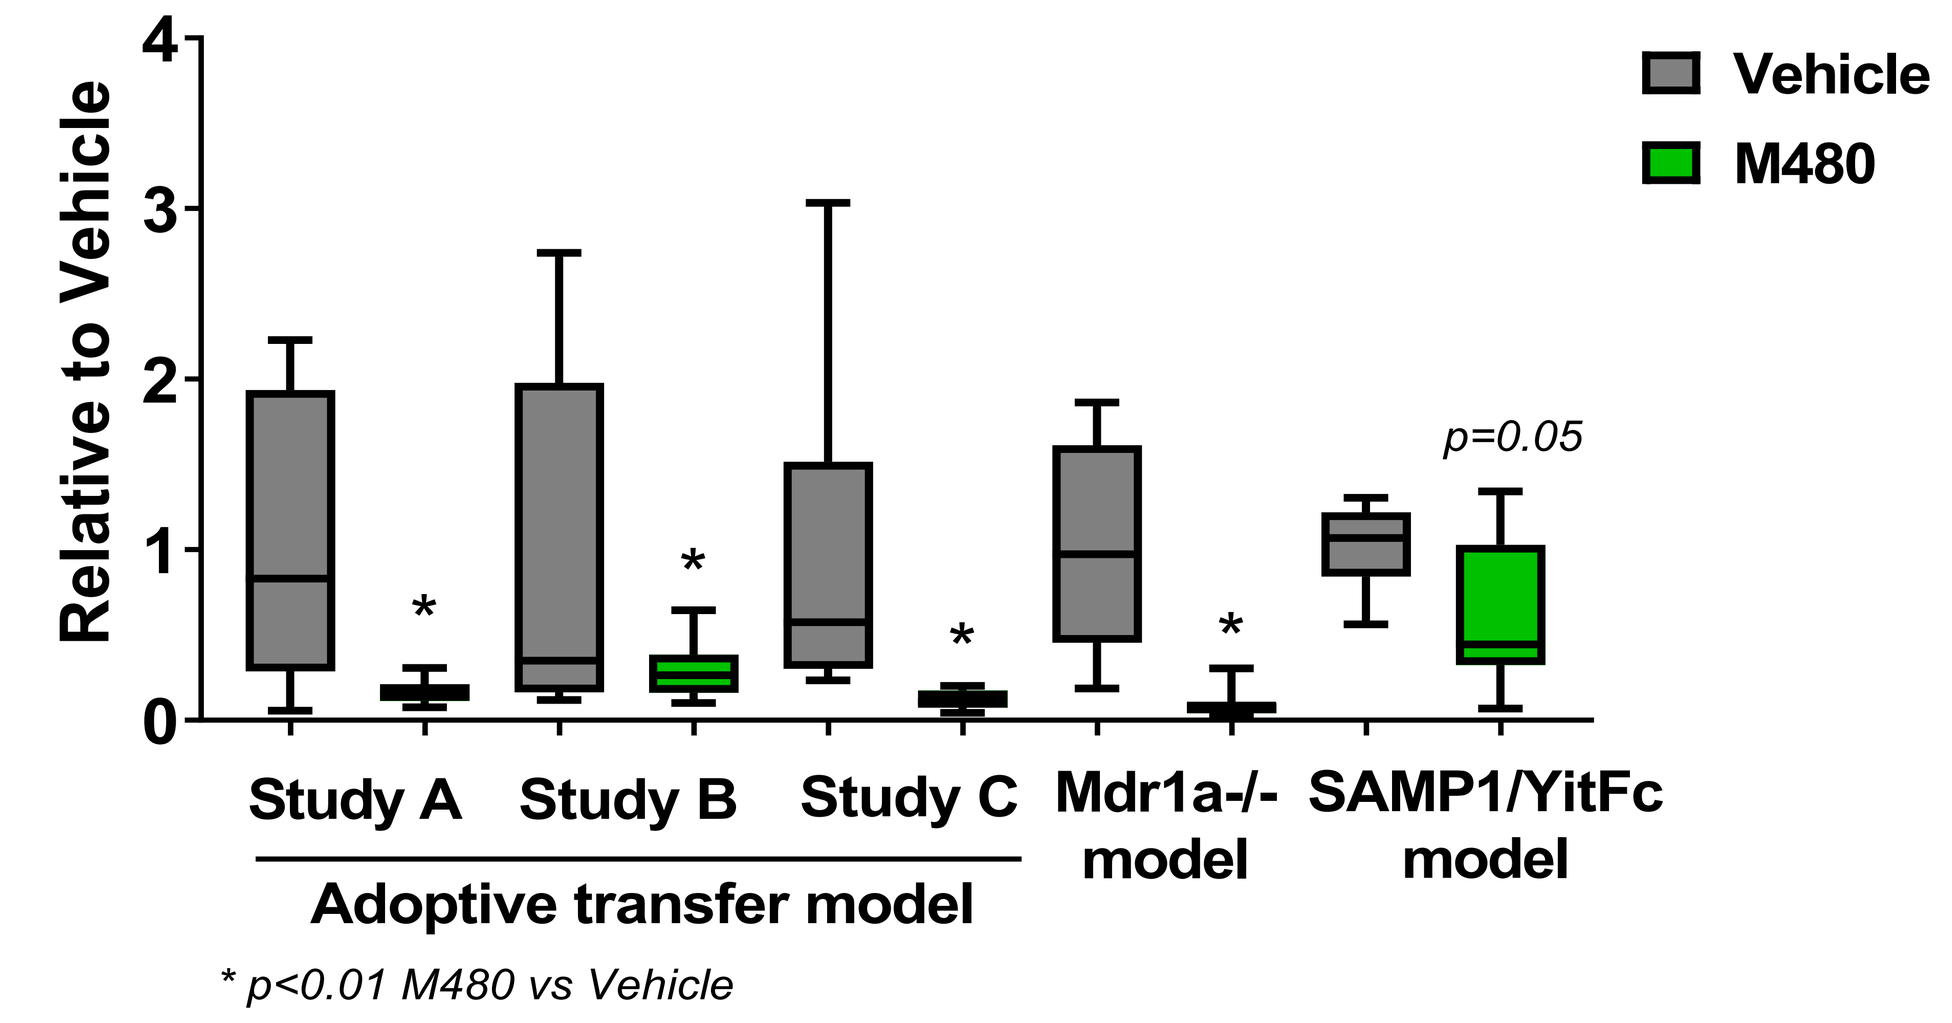

Relative to Vehicle
p=0.05
Vehicle
M480
Study A
Study B
Study C
Mdr1a-/- model
SAMP1/YitFc model
Adoptive transfer model

** p<0.01 M480 vs Vehicle*

Increased Anti-microbial Gene Expression in Multiple Models

Ang mRNA Expression

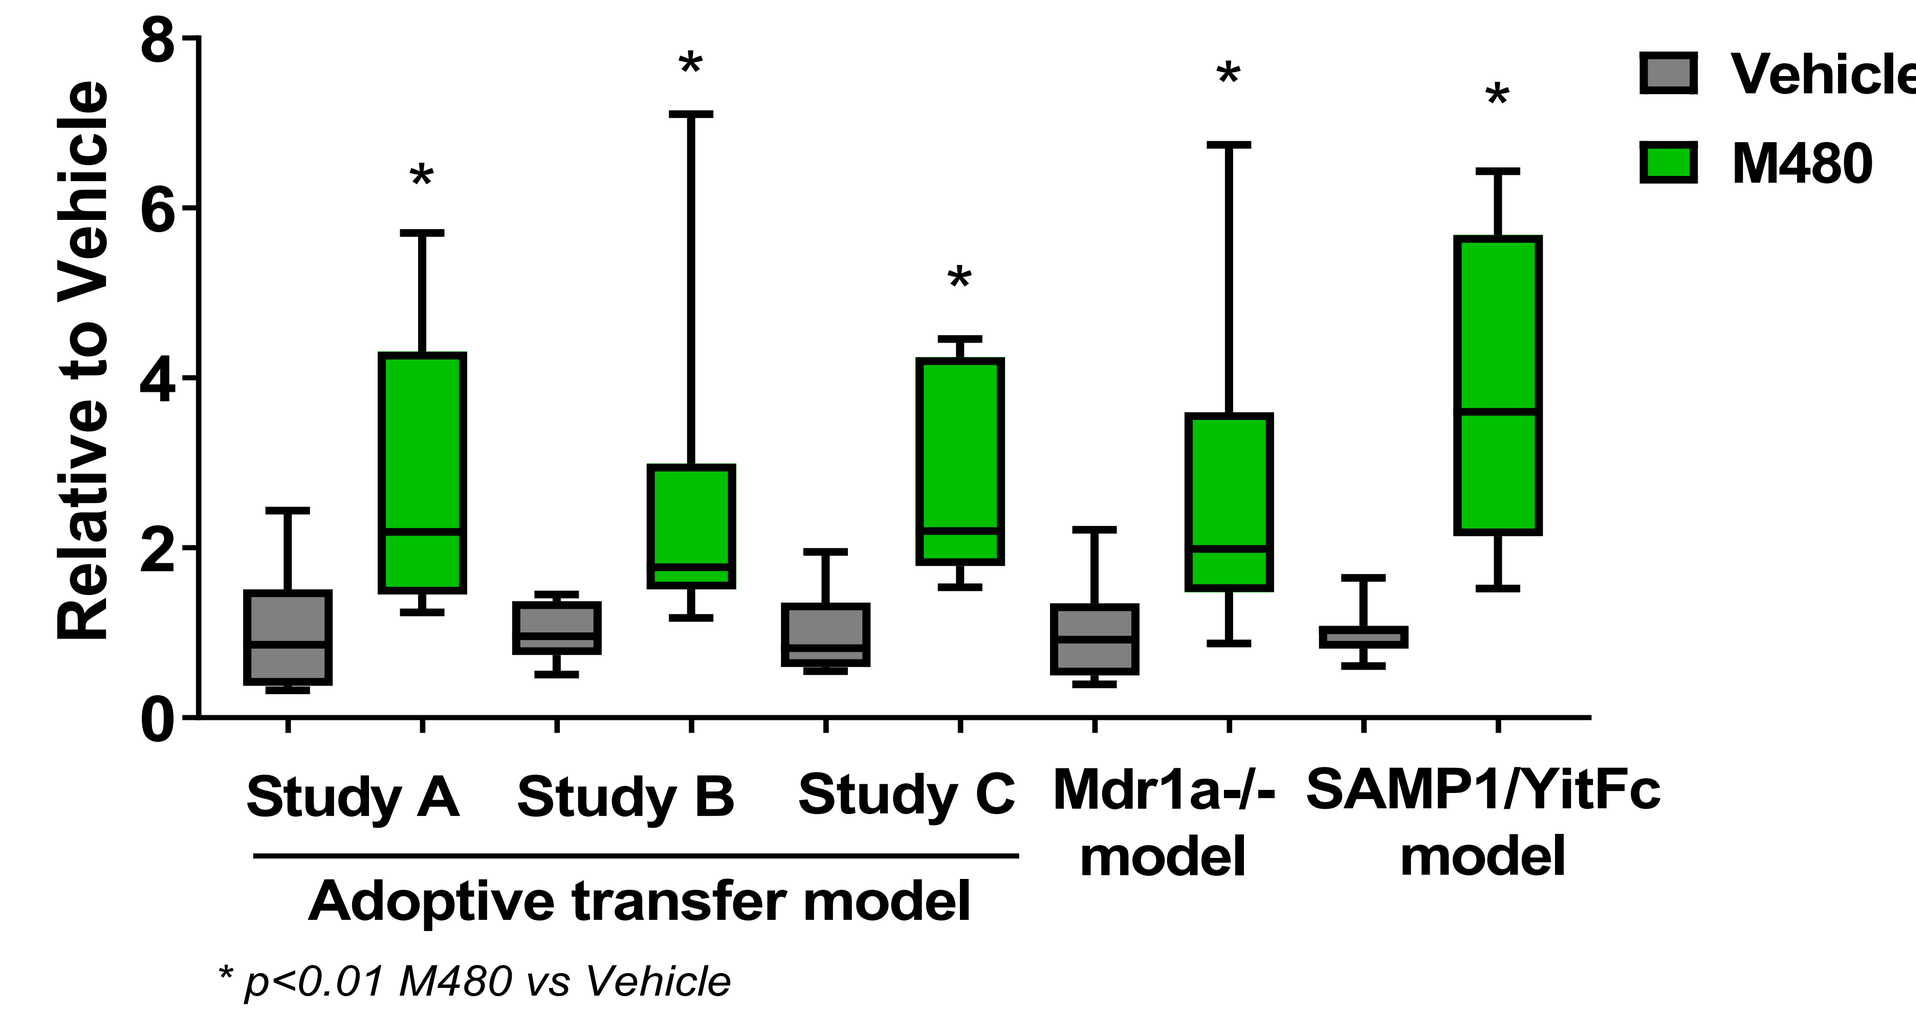

Relative to Vehicle
Vehicle
M480
Study A
Study B
Study C
Mdr1a-/- model
SAMP1/YitFc model
Adoptive transfer model

** p<0.01 M480 vs Vehicle*

NOS2 mRNA Expression

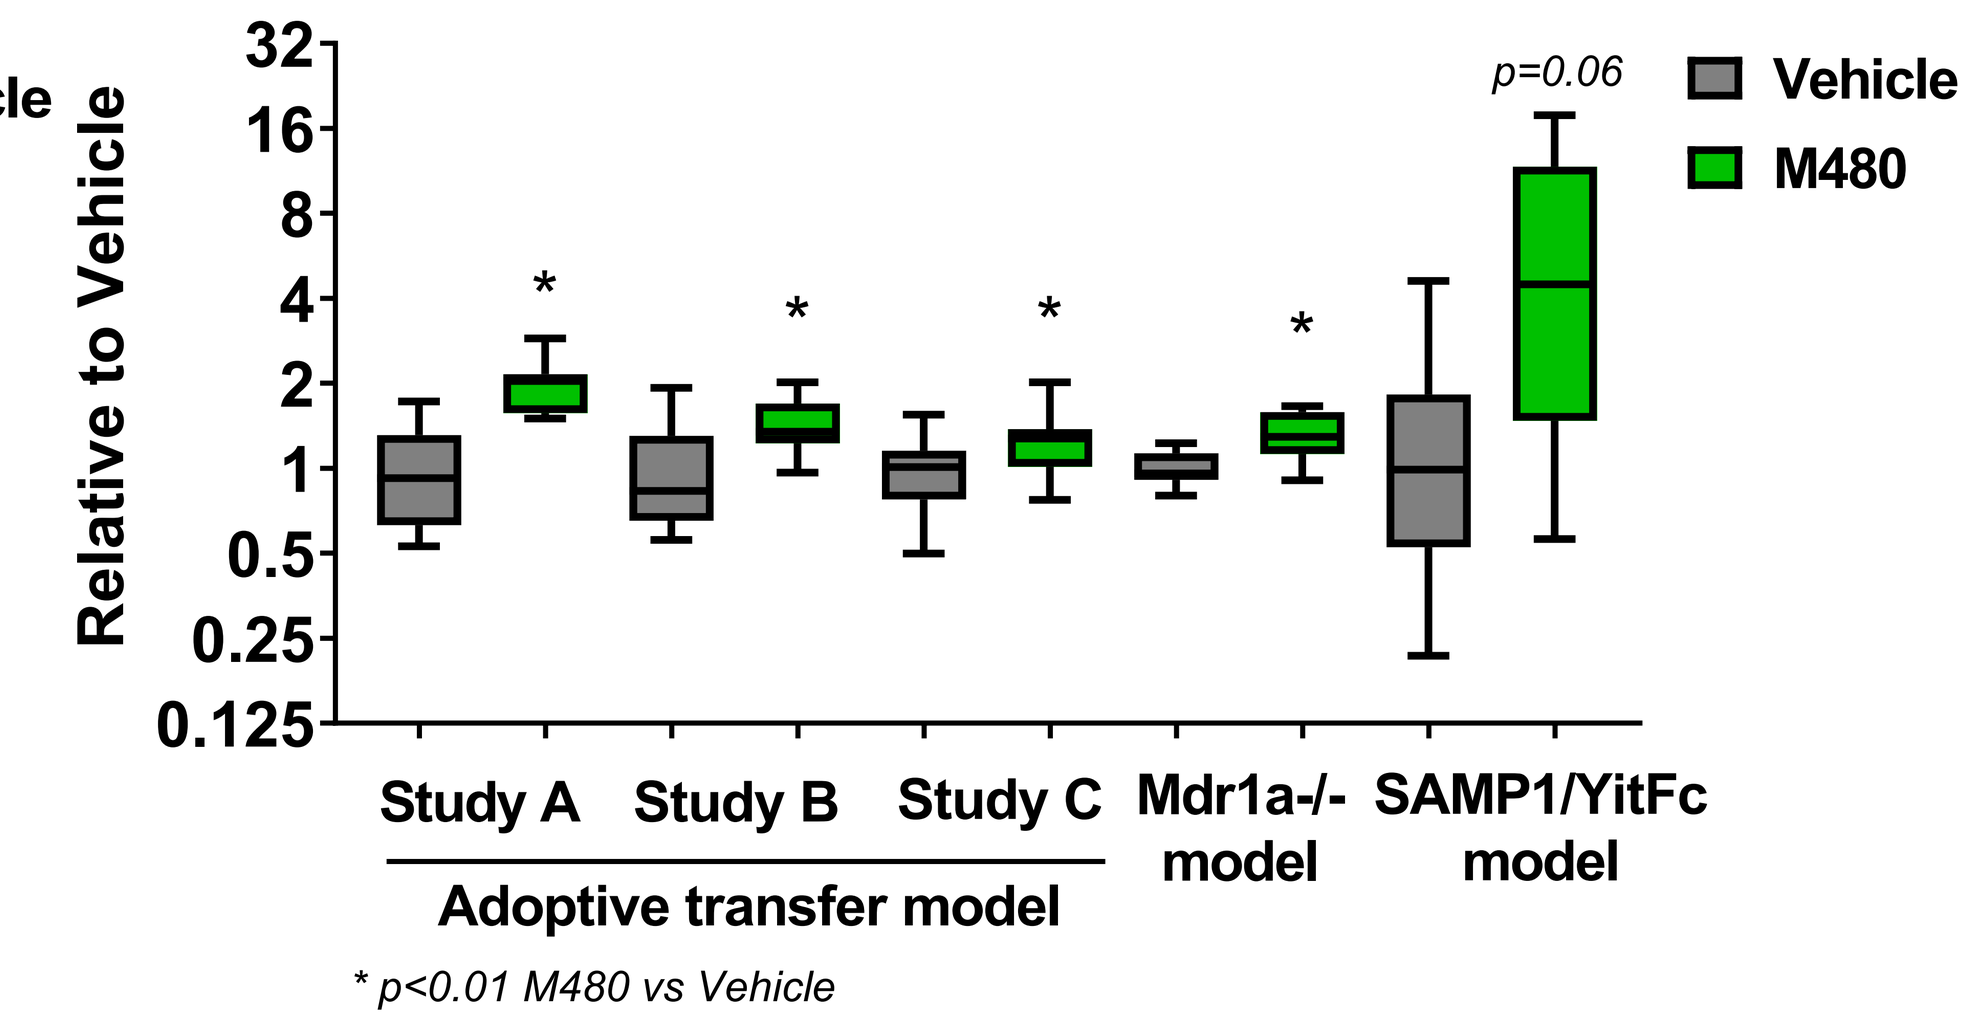

Relative to Vehicle
p=0.06
Vehicle
M480
Study A
Study B
Study C
Mdr1a-/- model
SAMP1/YitFc model
Adoptive transfer model

** p<0.01 M480 vs Vehicle*

Proposed Mechanism of Action for FXR Agonists in IBD

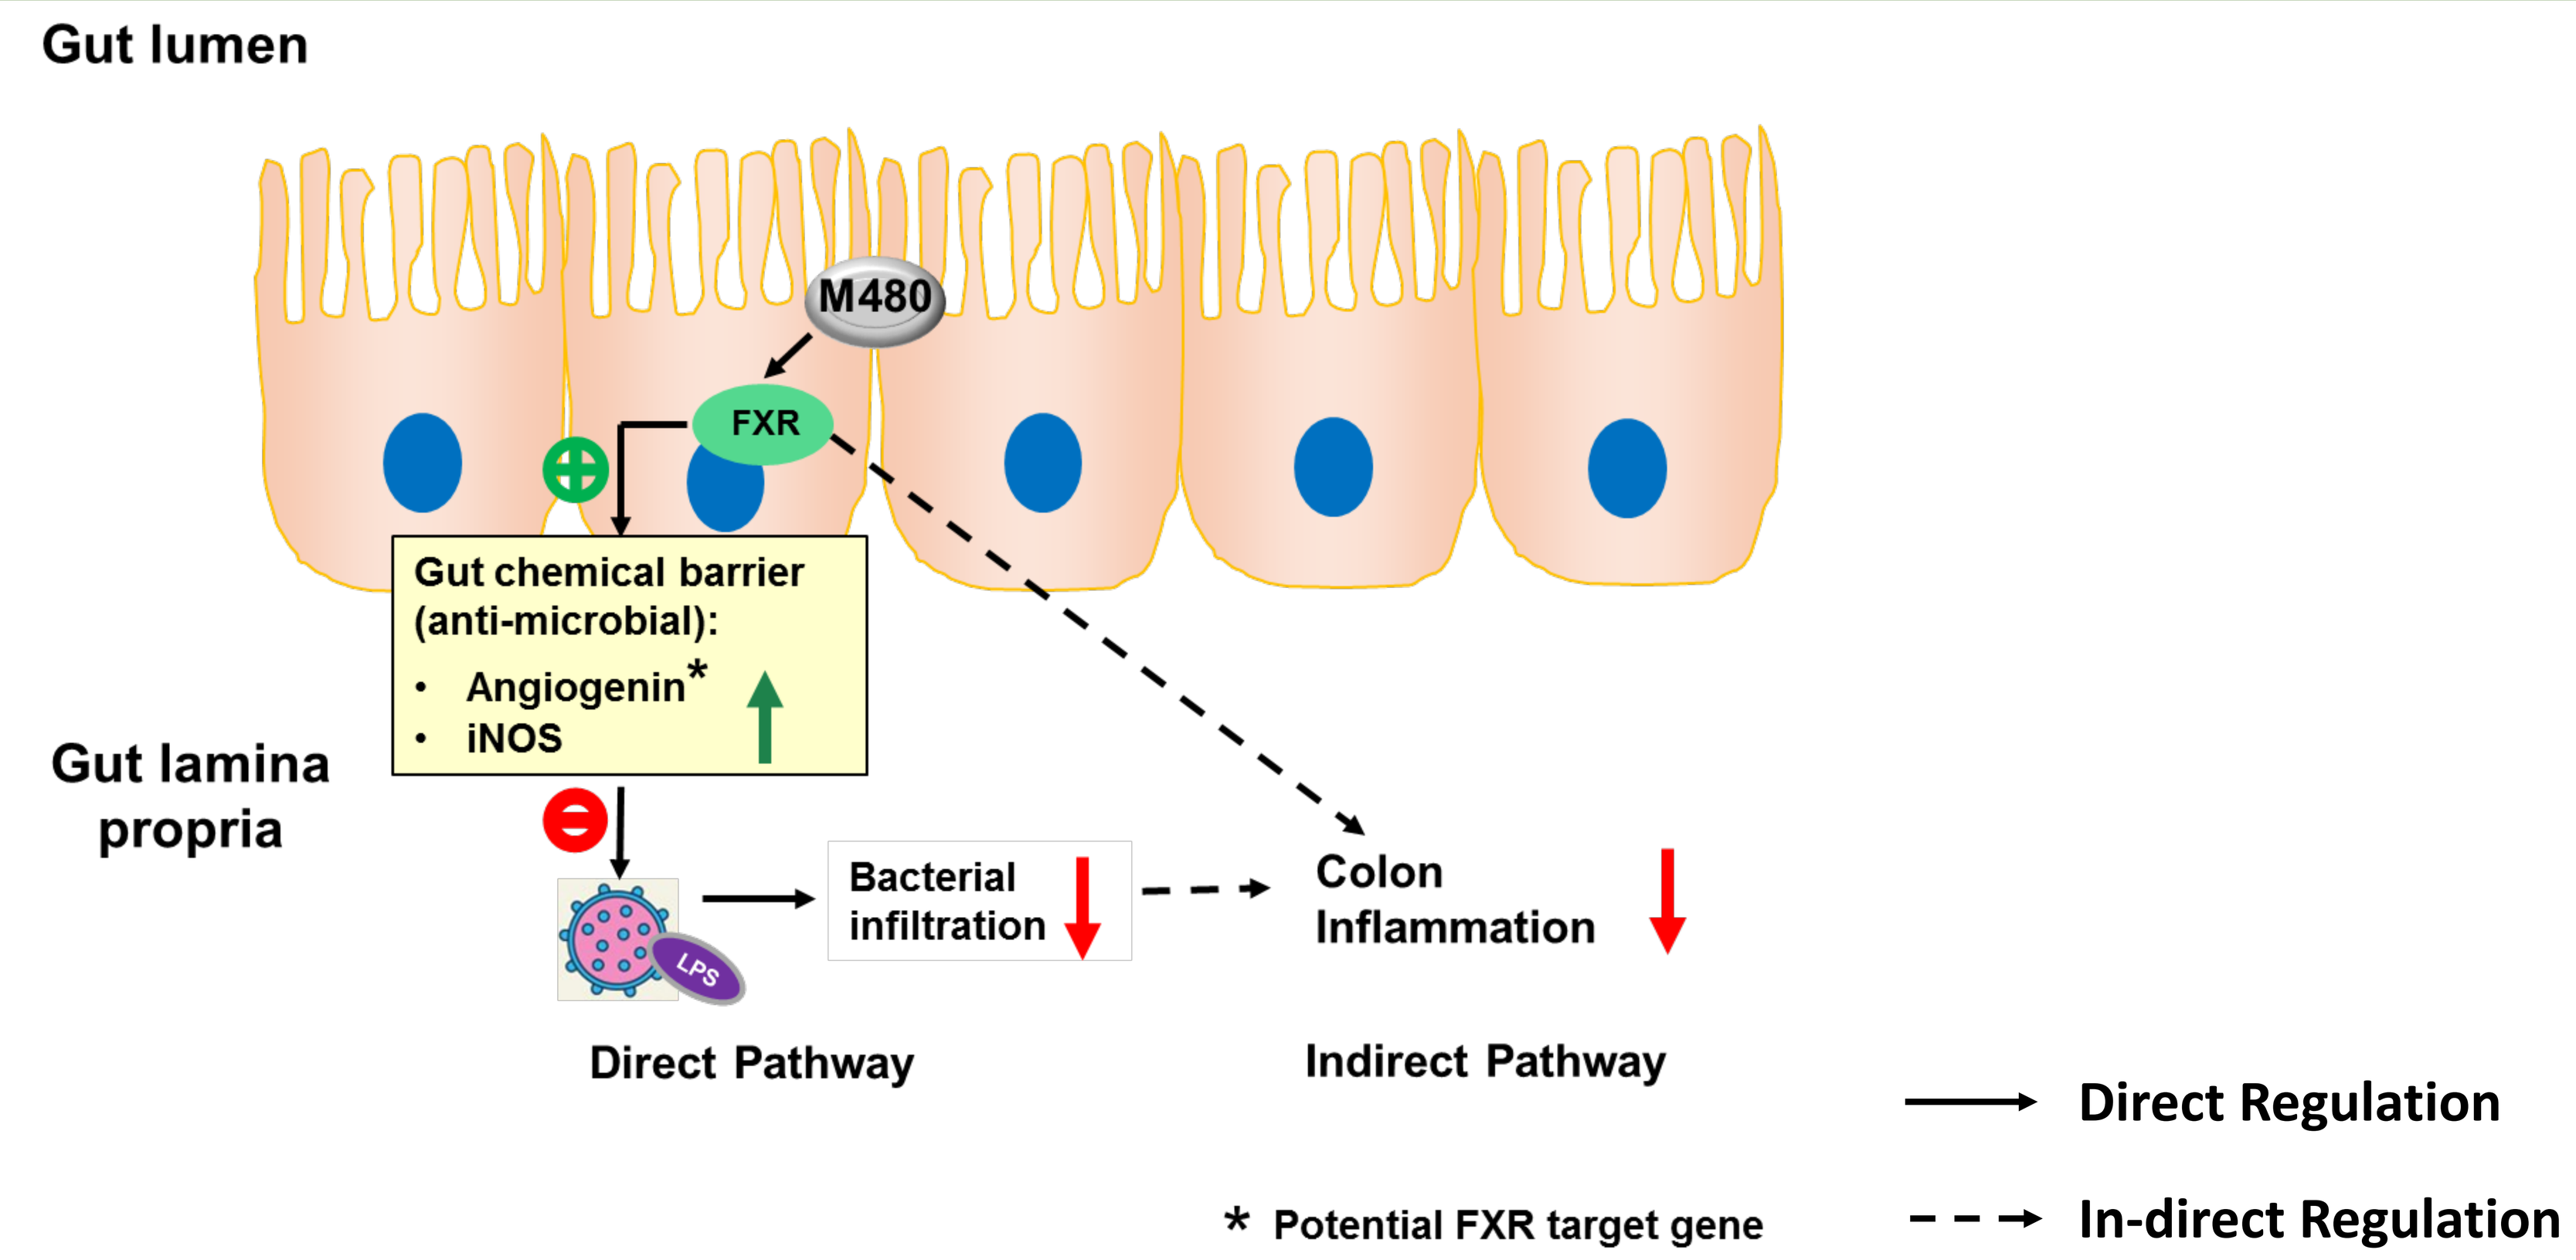

Gut lumen
M480
FXR
Gut chemical barrier (anti-microbial):
Angiogenin*
iNOS
Gut lamina propria
Bacterial infiltration
Colon Inflammation
Direct Pathway
Indirect Pathway
Direct Regulation
In-direct Regulation
* Potential FXR target gene

DISCLOSURES

B.W., S.G., J.N., K.D., H.D., A.M., K.L., A.O., J.Q., N.L., X.L., E.B., K.S. and N.S. are employees and equity holders in Metacrine, Inc.

Digestive Disease Week®