MET409, a Sustained FXR Agonist, Decreased Hepatic Fat and Improved Liver Function without Raising LDL-C after 28 Days in NASH Patients

Stephen A. Harrison[1], Mustafa R. Bashir[2], Jennifer Shim-Lopez[3], Ken Song[3], Hubert C. Chen[3]
[1]Pinnacle Clinical Research, San Antonio, Texas, USA; [2]Duke University Medical Center, Durham, North Carolina, USA; [3]Metacrine, Inc., San Diego, California, USA

METACRINE®
3985 Sorrento Valley Blvd., Suite C
San Diego, CA 92121 USA
www.metacrine.com

1. BACKGROUND

Activating farnesoid X receptor (FXR) has been clinically validated to improve non-alcoholic steatohepatitis (NASH) and fibrosis, although common drawbacks include moderate-severe pruritus within the therapeutic dose range.

Recently, non-bile acid agonists have shown limited efficacy with once-daily dosing, likely due to transient target engagement. Sustained FXR engagement may be key to an optimal therapeutic index based on preclinical and clinical studies.

MET409, an optimized FXR agonist with a novel non-bile acid structure and enhanced potency, has demonstrated sustained FXR engagement and favorable safety and tolerability profile after 14 days of dosing in healthy subjects.

2. OBJECTIVE

We conducted an open-label, single-center pilot study to determine the safety and pharmacodynamics (PD) of MET409 administered for 28 days in patients with NASH. Subjects were followed for an additional 28 days for safety observations.

3. METHODS

Men and women (18 to 75 years-old) with (1) biopsy-confirmed NASH or (2) transient elastography findings of kPa ≥8.5 and controlled attenuation parameter >300 dB/m were eligible for participation. All subjects had ≥ 10% liver fat content (LFC) by magnetic resonance imaging−proton density fat fraction at baseline.

Ten subjects received daily oral MET409 at 50 mg, which was shown to reduce plasma levels of 7α -hydroxy-4-cholesten-3-one (C4) by ~80% after 14 days in healthy subjects. The primary study objective was to assess the safety and tolerability of MET409. Secondary study objectives included liver function and LFC assessment.

4. RESULTS

MET409 at 50 mg was safe and well-tolerated. There were no serious adverse events, and all treatment-related adverse events were mild. No pruritus was reported by any of the subjects.

Liver Fat Reduction after 28 Days of Dosing

MET409 reduced LFC by -20.3% (mean relative change), with ≥30% relative reduction seen in 3 of 10 subjects.

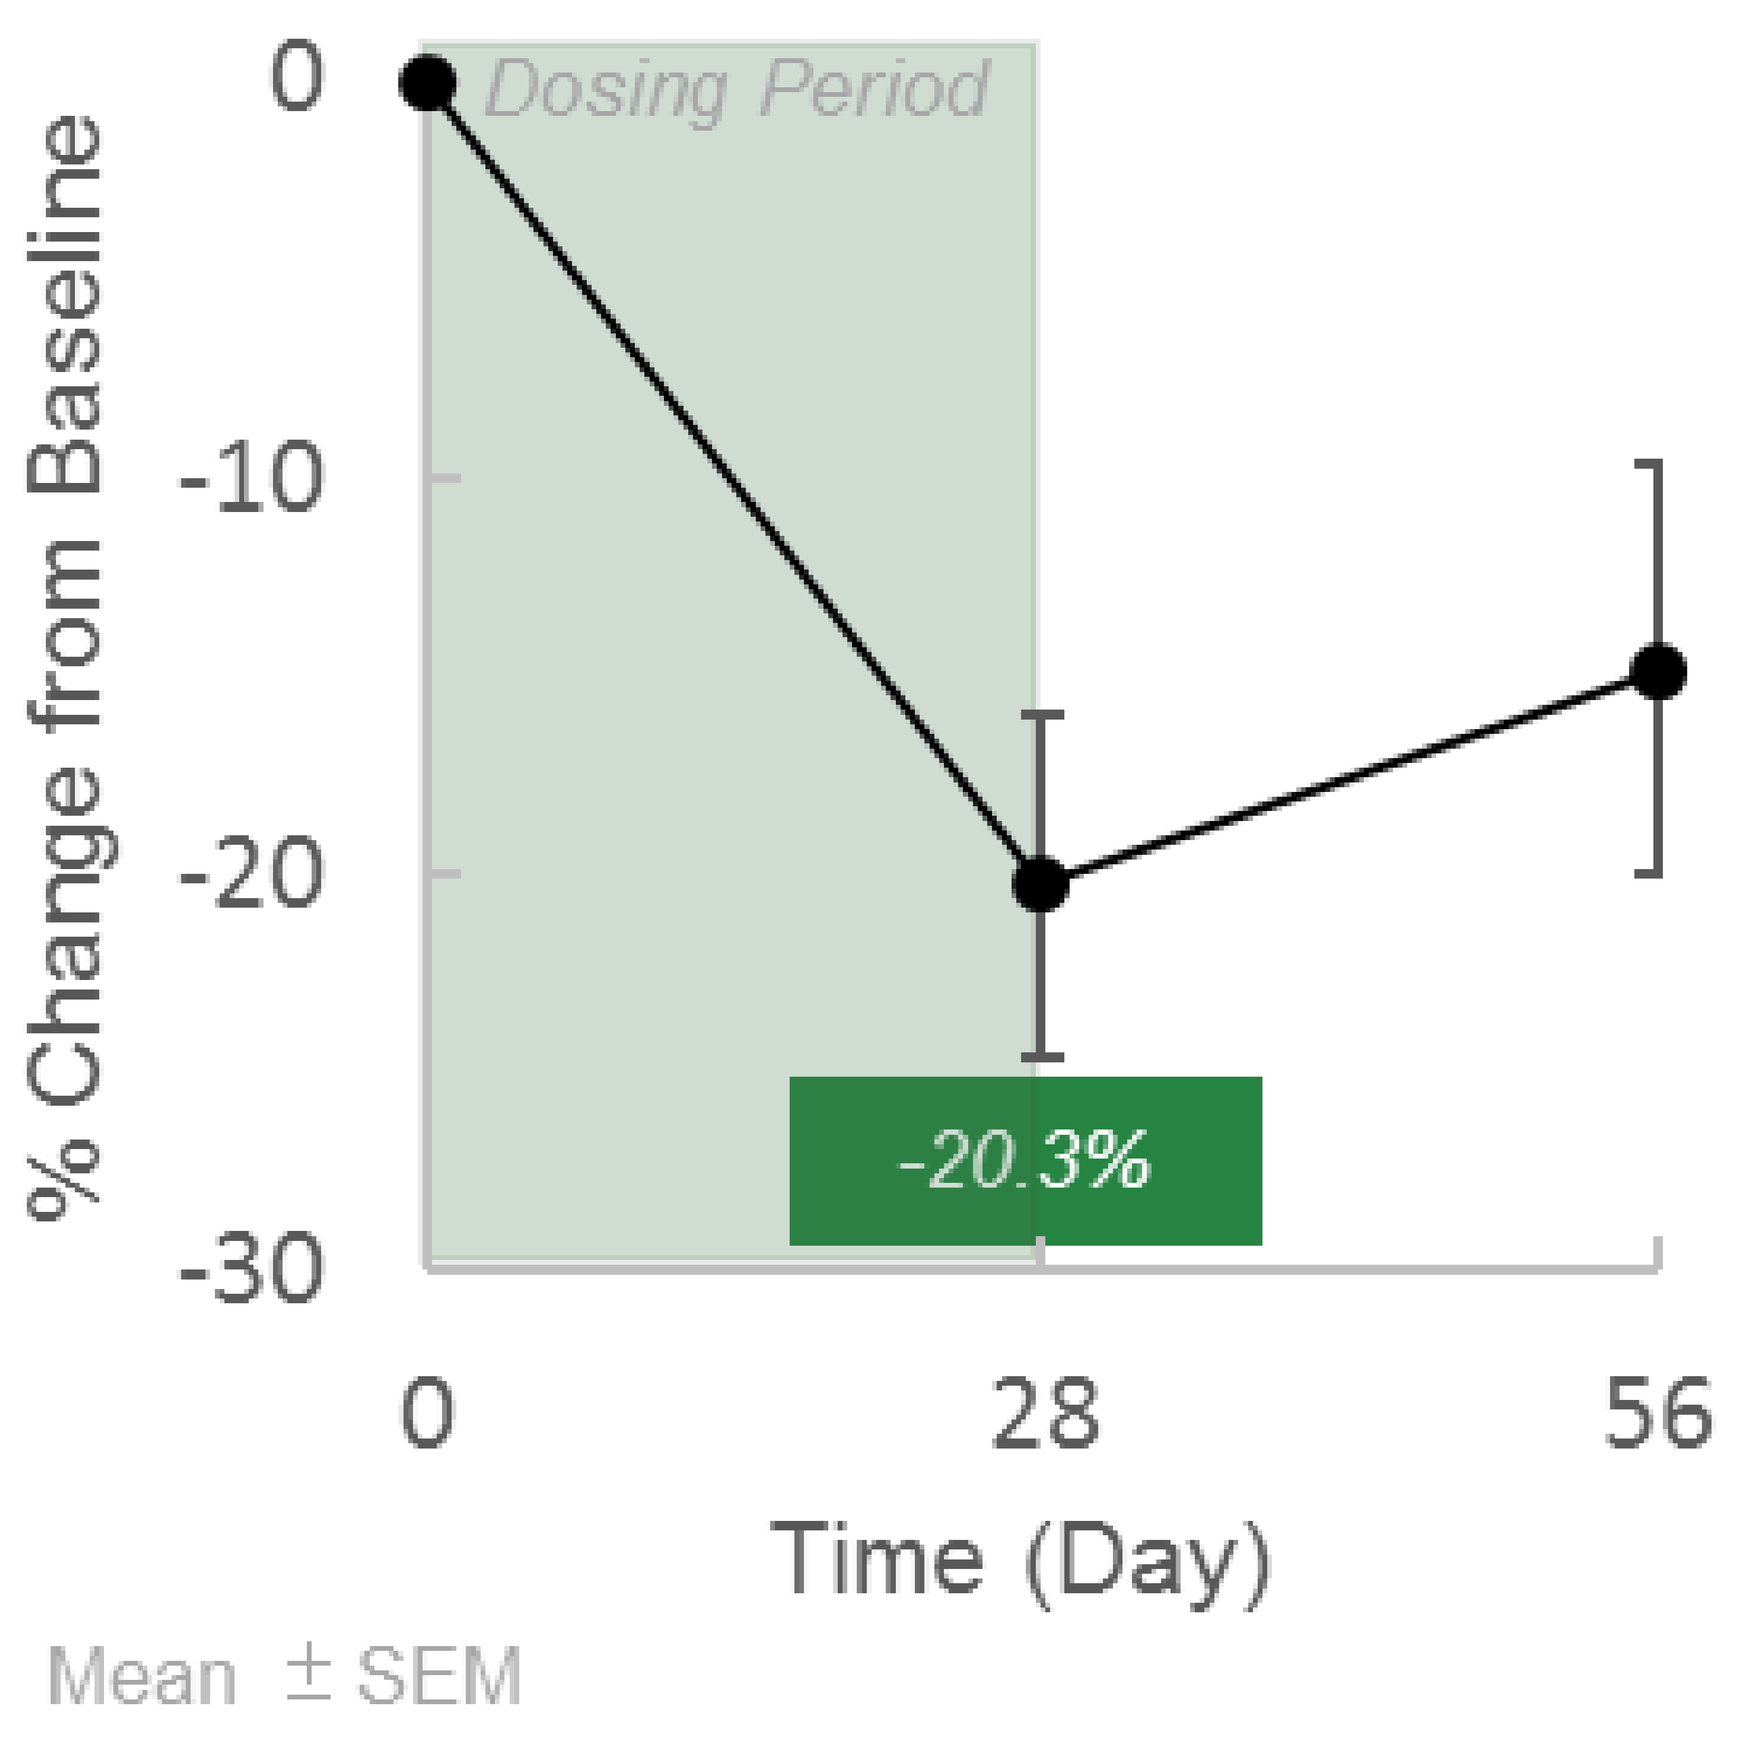
Dosing Period
% Change from Baseline
0
-10
-20
-30
-20.3%
0
28
56
Time (Day)
Mean ± SEM

Relative Change in Serum Lipids

	Day 14	Day 28
HDL-C	-6.7 ± 3.5%	-13.7 ± 4.0%
LDL-C	-0.2 ± 4.3%	-8.7 ± 5.0%
Triglycerides	-9.8 ± 6.4%	6.9 ± 6.9%

Mean ± SEM

MET409 resulted in an on-target reduction in high-density lipoprotein cholesterol (HDL-C). There were no meaningful changes in LDL-C or triglycerides.

Liver Fat Reduction Relative to Other FXR Agonists

The magnitude of reduction appears comparable to those observed with longer-term dosing of other FXR agonists.

Candidate	Profile/ Structure	Treatment Duration	Relative Liver Fat Reduction
MET409 (Metacrine)	Sustained/ Non-bile acid	4 wks	20% (low dose)
OCA (Intercept)	Sustained/ Bile acid	72 wks	17%*
Cilofexor (Gilead)	Transient, Non-bile acid	12 wks	8% (low dose) 16% (high dose)
Tropifexor (Novartis)	Transient, Non-bile acid	12 wks	7% (low dose) 31% (high dose)

*OCA is placebo subtracted

Time-Dependent Improvement in Liver Function

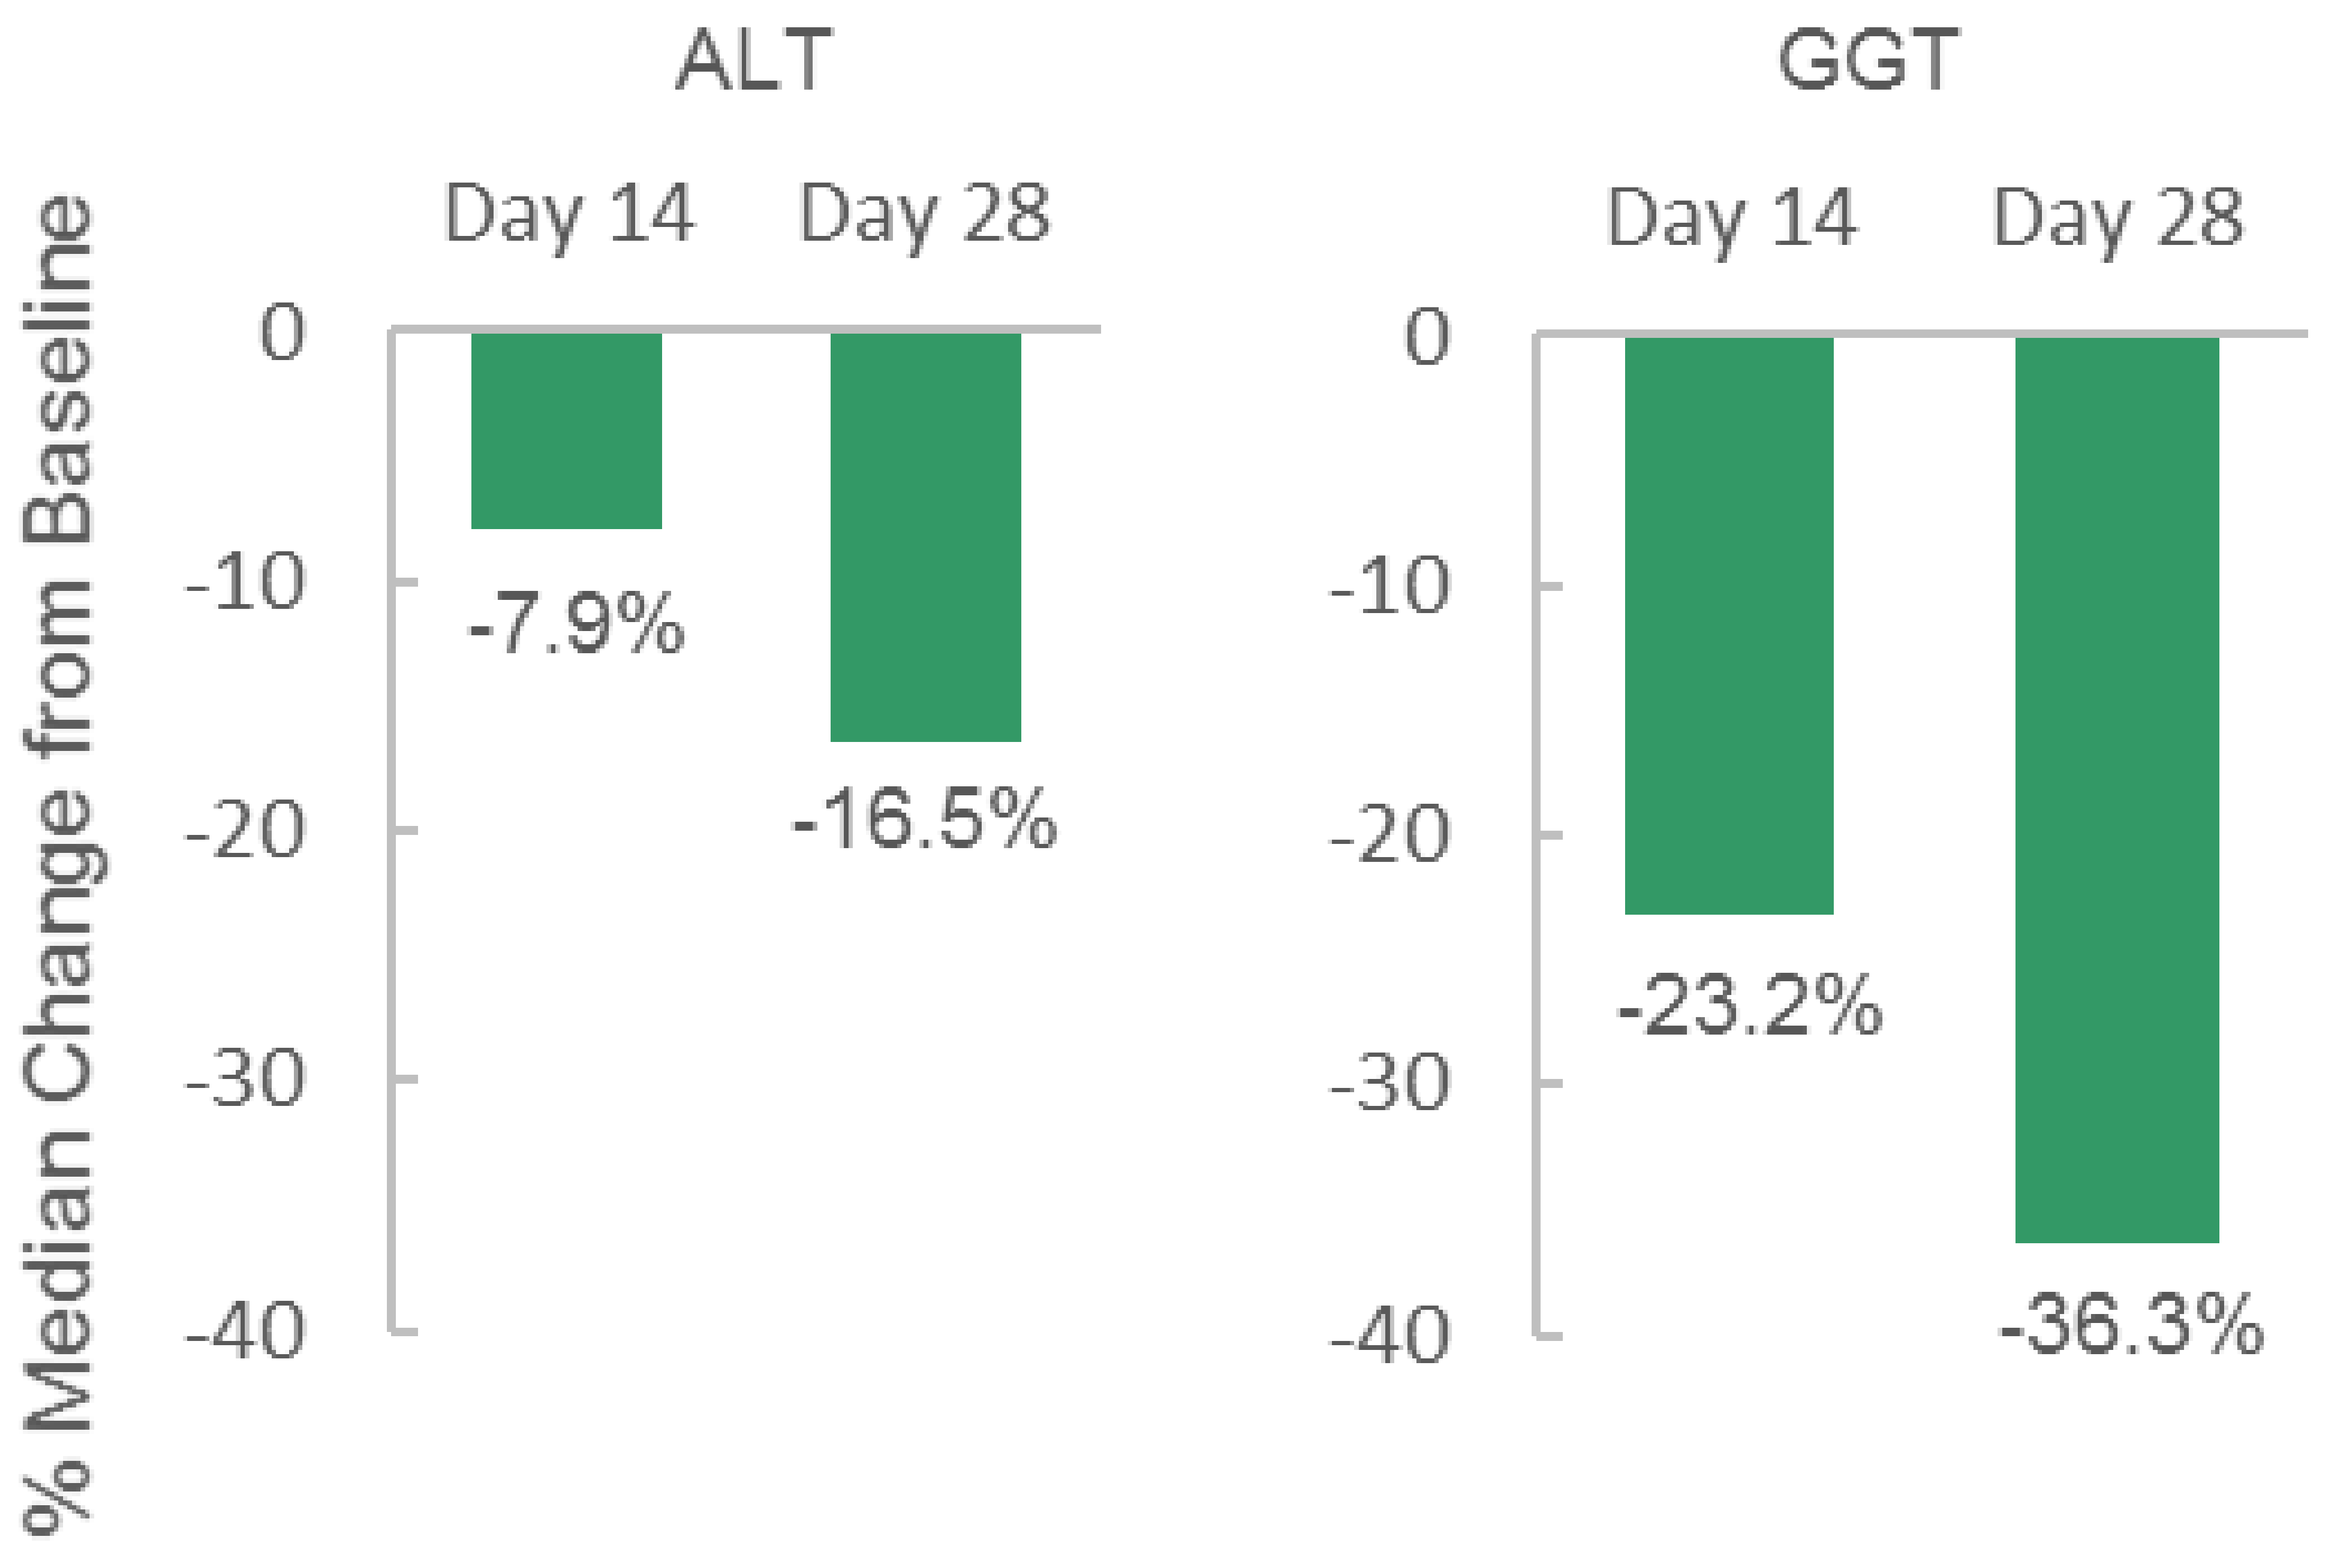
ALT
GGT
Day 14
Day 28
Day 14
Day 28
% Median Change from Baseline
0
-10
-20
-30
-40
-7.9%
-16.5%
-23.2%
-36.3%

MET409 lowered levels of alanine transferase (ALT) and gamma-glutamyl transferase (GGT) in a time-dependent manner.

5. CONCLUSION

In this open-label, single-center study, MET409 − an optimized sustained non-bile acid FXR agonist − reduced hepatic fat and improved liver function after 28 days of dosing in 10 NASH patients.

These results demonstrate the potential of Metacrine's sustained, non-bile acid FXR agonist to offer a wider therapeutic index and deliver a differentiated, best-in-class profile. Longer treatment duration with MET409 is being assessed in a placebo-controlled, multi-center study in NASH patients.